VALIDATION OF A SCORE FOR THE PREDICTION OF SERIOUS INFECTION IN PATIENTS WITH SYSTEMIC LUPUS ERYTHEMATOSUS: DATA FROM A LATIN AMERICAN LUPUS COHORT

GLADEL LATIN AMERICA

LUPUS 21-24 MAY 2025 TORONTO

Rosana Quintana; Guillermo Pons-Estel; Karen Roberts; Erika Susana Palacios Santillán; Iñigo Rúa-Figueroa; José M. Pego-Reigosa; Pablo Ibañez; Leonel A. Berbotto; María Constanza Bertolaccini; Marina Laura Micelli; Cecilia Nora Pisoni; Vitalina de Souza Barbosa; Henrique de Ataíde Mariz; Francinne Machado Ribeiro; Luciana P.C. Seguro; Emília Inoue Sato; Milena Mimica Davet; Gustavo Aroca Martinez; Fabio Bonilla-Abadía; Gerardo Quintana-López; Reyna Elizabeth Sánchez Briones; Mario Pérez Cristóbal; Luis H. Silveira; Ignacio García-De la Torre; Ivan Morales Avendaño; Pablo Gamez-Siller; Astrid Paats Nicora; Jorge Cieza-Calderon; Andy Armando Mendoza Maldonado; Martin Rebella; Gonzalo Silveira; John Fredy Jaramillo; Mónica Sánchez; Urbano Sbarigia; Ashley Orillion; Federico Zazzetti; Graciela S. Alarcón; and Bernardo Pons-Estel on behalf of the Grupo Latino Americano De Estudio del Lupus (GLADEL) .*

ID 447
*Presenting Author

Background

Patients with systemic lupus erythematosus (SLE) are at increased risk of serious infections, which in turn, are associated with morbidity and mortality. The Systemic Lupus Erythematosus Registry of the Spanish Society of Rheumatology (RELESSER) group has developed and internally validated a tool for prediction of serious infections in SLE, with a recently improved version (SLE SI Score Revised or SLESIS-R)1, being an accurate and reliable instrument. SLESIS-R includes age, previous SLE-related hospitalization, previous serious infection, and glucocorticoid dose. This study aimed to validate SLESIS-R in a multi-ethnic, multi-national Latin-American (LA) SLE cohort.

Methods

GLADEL 2.0 is an observational cohort from 10 LA countries of patients ≥18 years of age who fulfilled the 1982/1997 American College of Rheumatology (ACR) and/or the 2012 Systemic Lupus International Collaborating Clinics (SLICC) classification criteria. Patients with sufficient data at baseline and first annual visits were included. The outcome variable was any serious infection during the first year of follow up that led to hospitalization. Baseline demographics and clinical manifestations, disease activity (SLEDAI-2k), SLICC/ACR Damage Index (SDI) and treatments were examined. Logistic regression was used to examine the predictive effect of baseline variables on the development of serious infection in the first year of follow-up. Receiver operator characteristics (ROC) analysis was used to define the area under the curve (AUC) for SLESIS-R. The cut-off point with the best validity parameters (sensitivity and specificity) was identified.

Results

Patient characteristics
Of the 1016 patients who completed one-year follow-up, 208 (20.4%) had serious infections. Patients with serious infections were older, predominantly male, and had a longer disease duration **(Table 1)**. This group had more frequent general, cardiac, pulmonary, hematological and gastrointestinal involvement at baseline and had a higher SDI and higher proportion of previous hospitalization.

Factors associated with serious infection
Univariate and multivariate analyses **(Table 2)** show variables associated with serious infection: disease duration, pulmonary and gastrointestinal involvements, and baseline glucocorticoid use.

Validation of SLESIS-R
The AUC for the score was 0.922 (0.903-0.940) **(Figure 1)**. A score of 7 was chosen as the optimal cut-off point, demonstrating a sensitivity of 87% and specificity of 82%.

Conclusion

Almost a third of patients had serious infections during the first year of follow-up. The score performed well in predicting serious infections, similar to the original score.

TABLE 1.
Comparison between groups according to their baseline clinical characteristics, disease activity, damage index, and treatments

	SERIOUS INFECTION No (N=808)	SERIOUS INFECTION Yes (N=208)	*p*-value	TOTAL (n=1016)
Age, Median [Q1, Q3]	34.7 [27.1, 44.2]	37.0 [28.2, 45.6]	**0.068**	35.3 [27.2, 44.3]
Female gender, n(%)	733 (90.7)	177 (85.1)	**0.025**	910 (89.6)
Disease duration (years), Median [Q1, Q3]	4.9 [1.3, 10.9]	9.3 [3.0, 15.2]	**<0.001**	5.6 [1.6, 11.7]
Ethnic Group, n(%)				
Afro-Latin American	68 (8.4)	15 (7.2)	0.621	83 (8.1)
Indigenous	5 (0.6)	3 (1.4)		8 (0.7)
Mestizo	537 (66.5)	133 (63.9)		670 (65.9)
Other	2 (0.2)	0 (0)		2 (0.1)
Caucasian	193 (23.9)	56 (26.9)		249 (24.5)
Baseline clinical features, n(%)				
General involvement	602 (75.1)	181 (87.0)	**<0.001**	783 (77.5)
Cutaneous involvement	726 (90.1)	194 (93.3)	0.200	920 (90.7)
Articular involvement	671 (83.4)	173 (83.2)	1.000	844 (83.3)
Hematological involvement	644 (80.1)	180 (86.5)	**0.042**	824 (81.4)
Renal involvement	479 (59.4)	131 (63.0)	0.383	610 (60.1)
Cardiac involvement	88 (10.9)	41 (19.8)	**<0.001**	129 (12.7)
Pulmonary involvement	56 (6.9)	34 (16.4)	**<0.001**	90 (8.8)
Gastrointestinal involvement	94 (11.7)	39 (18.8)	**0.009**	133 (13.1)
Neurological involvement	11 (1.3)	2 (0.9)	1.000	13 (1.28)
Serosal involvement	254 (31.6)	69 (33.2)	0.724	323 (31.9)
Hypocomplementemia*	649 (80.3)	178 (85.6)	0.102	827 (81.4)
SLEDAI, Median [Q1, Q3]	4.0 [1.0, 10.0]	6.0 [2.0, 12.0]	0.274	5.0 [1.0, 11.0]
SDI, Median [Q1, Q3]	0 [0, 1.0]	1.00 [0, 2.0]	**<0.001**	0 [0, 1.0]
Previous SLE-related hospitalization	486 (60.4)	208 (100)	**<0.001**	694 (68.6)
Previous serious infection	359 (44.4)	187 (89.9)	**<0.001**	546 (53.7)
Baseline treatments, n(%)				
Glucocorticoids			0.216	
≤5 mg	193 (34.8)	44 (27.3)		237 (33.1)
>5 mg and<10 mg	99 (17.9)	37 (23.0)		136 (19.0)
≥10 mg and<30 mg	146 (26.4)	48 (29.8)		194 (27.1)
≥30 mg	116 (20.9)	32 (19.9)		148 (20.7)
Antimalarials	780 (97.6)	199 (95.7)	0.198	979 (97.2)
Cyclophosphamide-IV	76 (11.6)	24 (13.0)	0.094	100 (11.9)
Mycophenolate	269 (40.6)	79 (42.2)	0.074	348 (40.9)
Azathioprine	117 (17.8)	26 (14.0)	**<0.001**	143 (16.9)
Rituximab	29 (4.4)	9 (4.8)	0.205	38 (4.5)
Belimumab	14 (2.1)	5 (2.6)	0.809	19 (2.2)

*At least one of the following C3 or C4 or CH50; SLEDAI: Systemic Lupus Erythematosus Disease Activity Index; SDI: Systemic Lupus International Collaborating Clinics/American College of Rheumatology Disease Index; Glucocorticoids expressed as mg of prednisone per day.

TABLE 2.
Univariate and multivariate analysis of serious infection in SLE GLADEL patients

	UNIVARIATE OR (95% CI)	*p*-value	*Multivariate OR (95% CI)*	*p*-value
Age (years)≥60	1.1(0.5-2.2)	0.600		
Male gender	1.7(1.1-2.6)	0.019		
Disease duration	1.0(1.1-1.2)	<0.001	1.1(1.1-1.2)	<0.001
Hematological involvement	1.6(1.0-2.5)	0.035		
Cardiac involvement	2.0(1.3-3.0)	<0.001		
Pulmonary involvement	2.6(1.6-4.1)	<0.001	2.3(1.4-3.7)	<0.001
Gastrointestinal involvement	1.7(1.1-2.6)	<0.007	1.5(1.0-2.4)	0.033
General involvement	2.2(1.4-3.5)	<0.001		
Hypocomplementemia	1.4(0.9-2.2)	0.084		
SDI	1.4(1.2-1.6)	<0.001		
GC ≥30 mg at baseline	1.3(0.8-2.0)	0.200	1.5(1.1-2.4)	0.038
Azathioprine	1.1(0.6-1.7)	0.700		
Cyclophosphamide-IV	1.1(0.6-1.9)	0.600		

OR: odd ratio; CI: confidence interval; SDI: Systemic Lupus International Collaborating Clinics/American College of Rheumatology Disease Index; GC: glucocorticoids expressed as mg of prednisone/day.

FIGURE 1.
ROC curve for the SLESIS-R score.

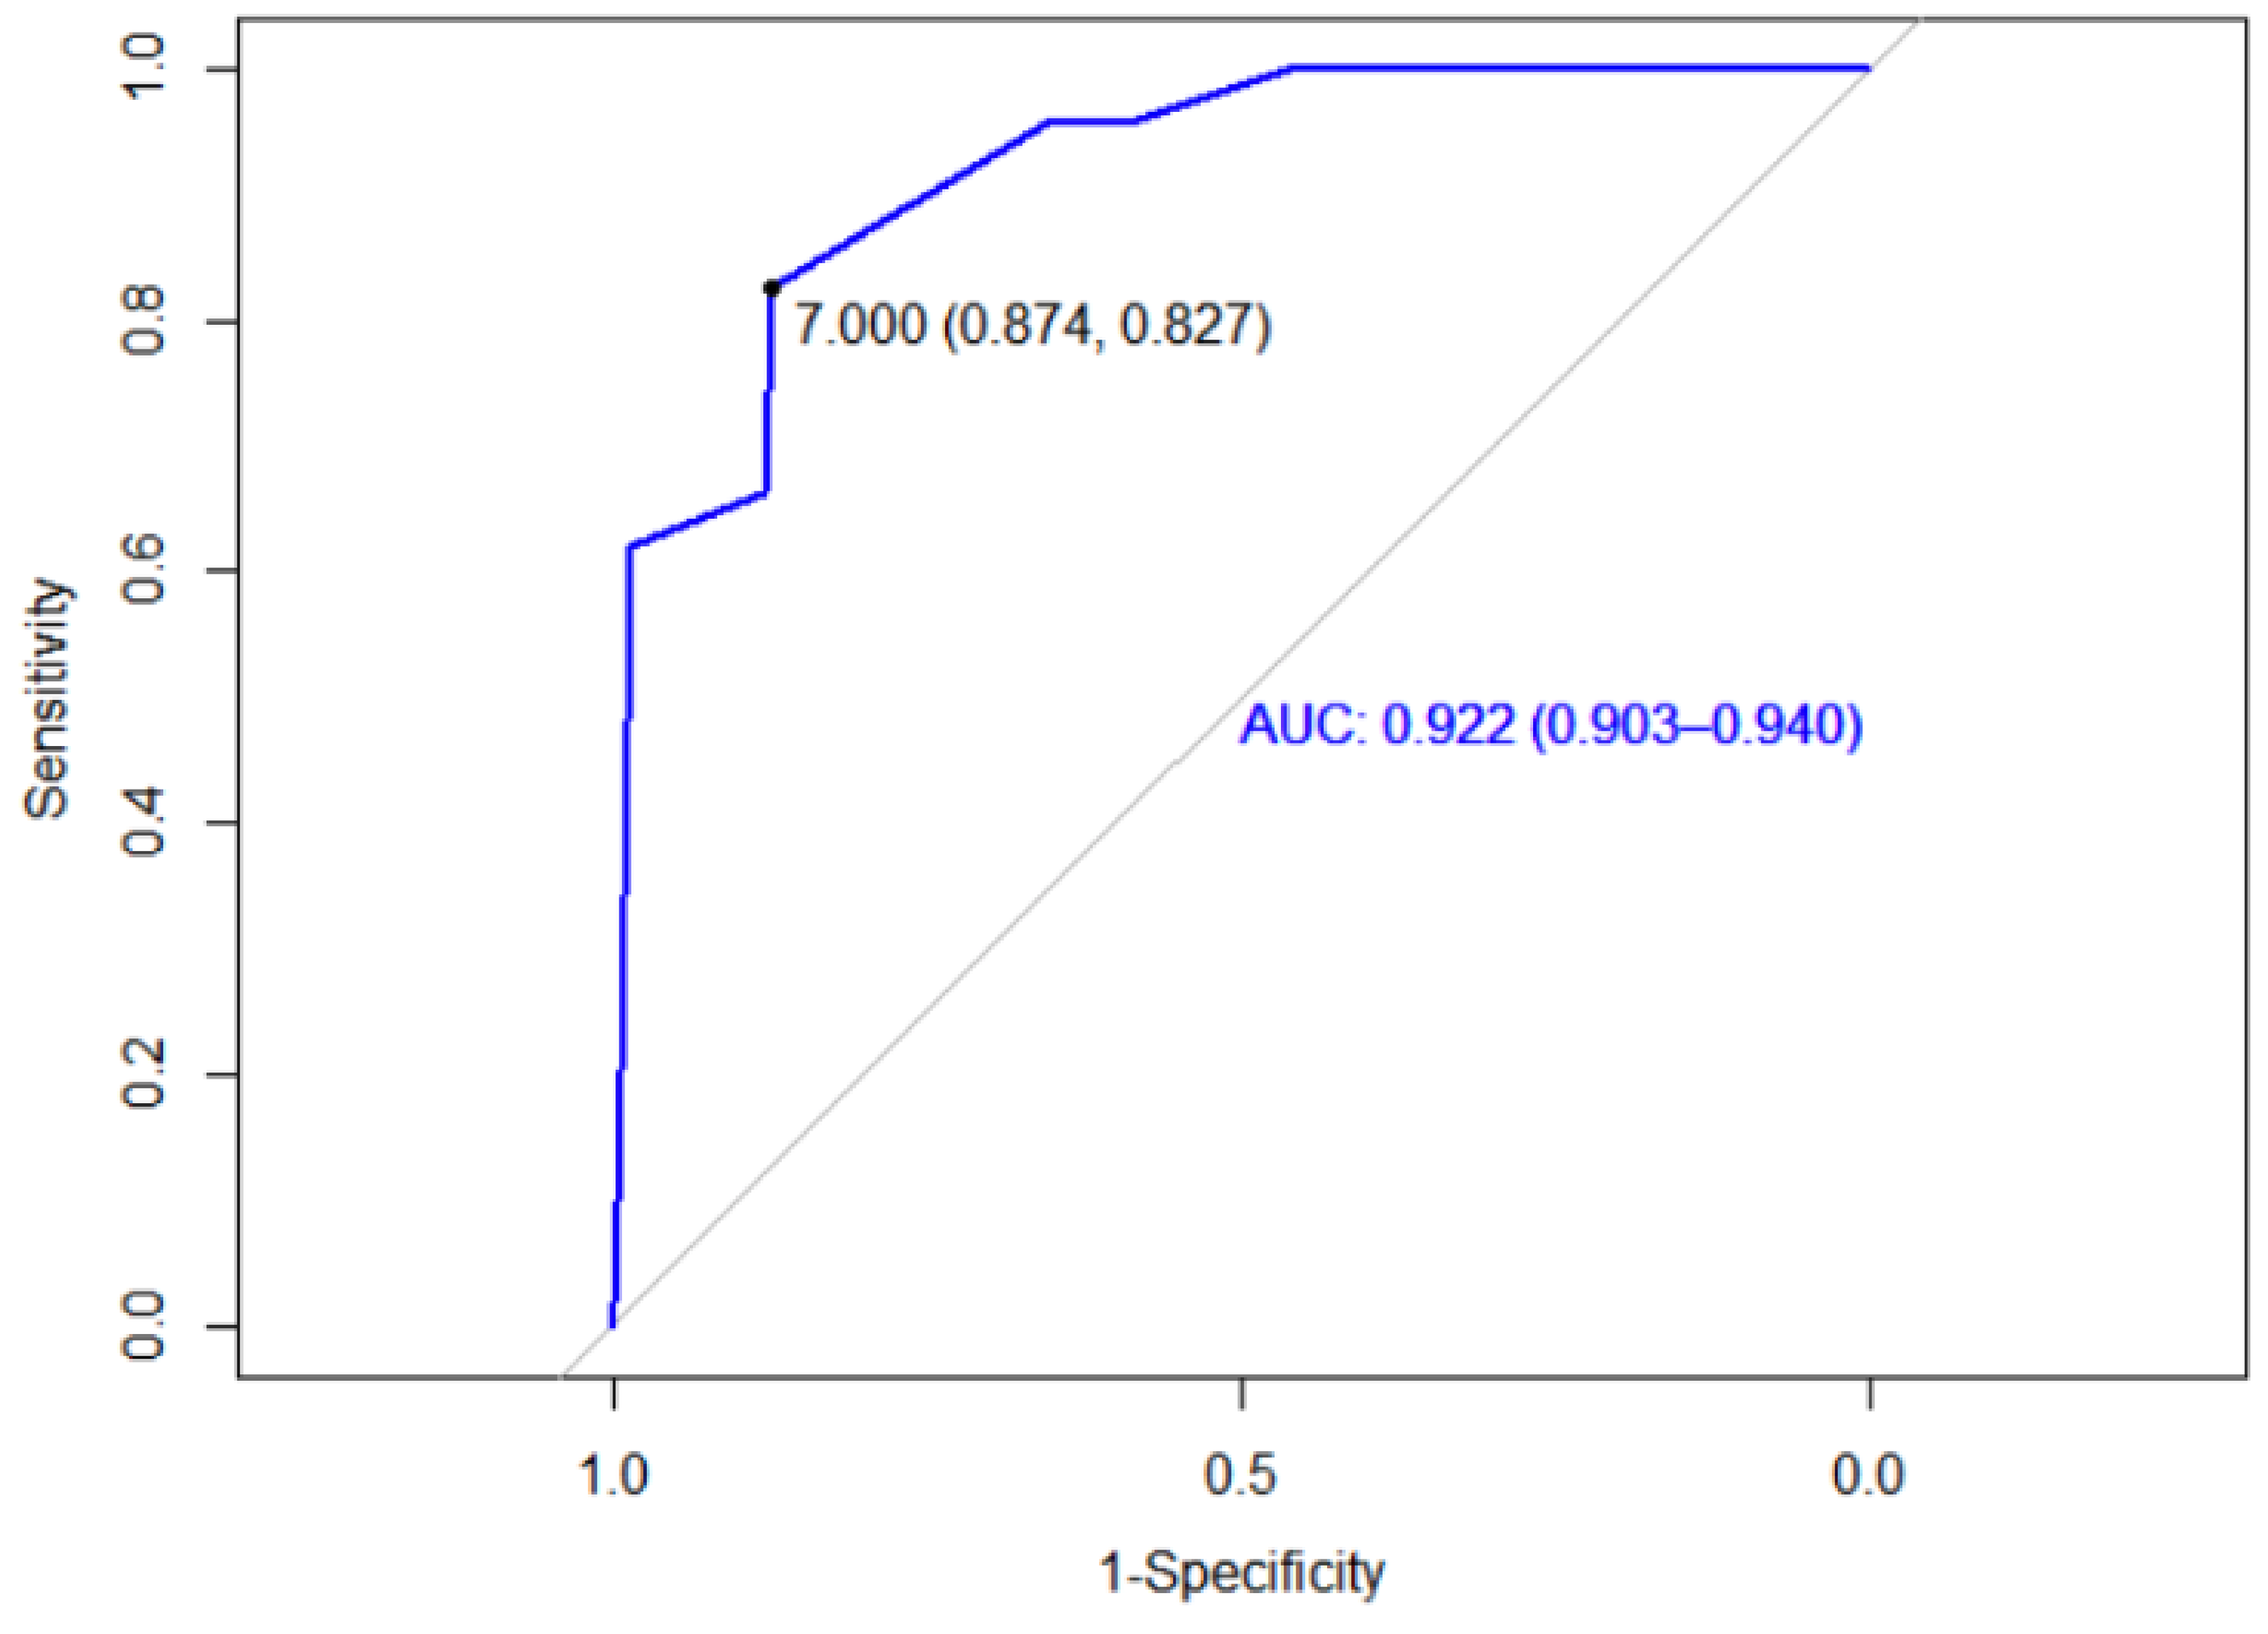
Sensitivity
1-Specificity
0.0
0.2
0.4
0.6
0.8
1.0
1.0
0.5
0.0
7.000 (0.874, 0.827)
AUC: 0.922 (0.903–0.940)

ROC: receiver operator characteristics; SLESIS-R: systemic lupus erythematosus serious infection score revised; AUC: area under the curve.

References

1. Rua-Figueroa I, et al. Lupus Sci Med. 2024;11:e001096. doi:10.1136/lupus-2023-001096

Acknowledgments

This study was sponsored by Janssen Research & Development, LLC. Editorial support was provided by Panita M. Trenor, PhD, of Lumanity Communications Inc., and was funded by Janssen Global Services, LLC.