1530

THE RENAL ACTIVITY INDEX FOR LUPUS IDENTIFIES AND PREDICTS COMPLETE RENAL REMISSION OR ABSENCE OF KIDNEY INVOLVEMENT IN SYSTEMIC LUPUS ERYTHEMATOSUS

Guillermo Pons-Estel*, Rosana Quintana, Romina Nieto, Hermine Brunner, Marina Scolnik, Carmen Funes Soaje, Paula Alba, Verónica Saurit, Mercedes Argentina García, Guillermo A. Berbotto, Verónica Bellomio, Eduardo Mario Kerzberg, Graciela Gómez, Cecilia Pisoni, Vicente Juarez, Ana Malvar, Nílzio Antonio da Silva, Odirlei André Monticielo, Henrique Ataide Mariz, Francinne Machado Ribeiro, Eduardo F. Borba, Eloisa Bonfa, Edgard Torres dos Reis Neto, Iris Guerra Herrera, Loreto Massardo, Gustavo Aroca Martinez, Lorena Gómez Escorcia, Carlos A. Cañas Davila, Gerardo Quintana-Lopez, Carlos Enrique Toro-Gutierrez, Mario Moreno Alvarez, Miguel A. Saavedra Salinas, Margarita Portela Hernandez, Hilda Fragoso Loyo, Luis H. Silveira, Ignacio García-De La Torre, Carlos Abud Mendoza, Jorge Antonio Esquivel-Valerio, Isabel Acosta Colman, Astrid Paats, Claudia Mora Trujillo, Manuel F. Ugarte-Gil, Armando Calvo Quiroz, Roberto Muñoz Louis, Martin Rebella, Álvaro Danza, Federico Zazzetti, Ashley Orillion, Bernardo Pons-Estel on behalf of Grupo Latino Americano De Estudio de Lupus (GLADEL).

*Presenting author

BACKGROUND

- Effective, non-invasive disease activity and treatment response assessments are needed for patients with systemic lupus erythematosus (SLE), especially if associated with kidney disease, i.e. lupus nephritis (LN)
- The treatment goal of LN is the achievement of complete renal remission (CRR), and otherwise unexplained proteinuria of >0.5 gram which can prompt a kidney biopsy to diagnose LN
- The Renal Activity Index for Lupus (RAIL) measures the degree of kidney inflammation
- The RAIL-score is calculated from the creatinine-adjusted RAIL biomarkers (NGAL, KIM-1, MCP-1, adiponectin, hemopexin, ceruloplasmin)[1]
 - Higher scores indicate higher kidney inflammation

OBJECTIVE

- Evaluate the role of RAIL to distinguish CRR status and identify the cut-point for RAIL
- Evaluate the role of RAIL in predicting change in CRR status over time

METHODS

- Urine samples collected from 69 SLE adult patients with and without LN were studied longitudinally at enrollment into the GLADEL cohort (T0), at 6 months (T1) and 12 months (T2)
- Absolute scores and changes in RAIL-scores over time were assessed for achievement of CRR status [proteinuria of <0.5 grams] by logistical regression models
- The Youden Index optimal cut-points on the receiver operating characteristic (ROC) curves were calculated

RESULTS

- Patient characteristics at enrollment and disease course at 6 months and 12 months are shown for 51 patients with LN (74%; 91% female) and 18 (26%) patients without a diagnosis of LN over a total of 186 visits (**Table 1**)
- RAIL-scores were correlated with renal-SLEDAI (r=0.46; P<0.0001) and proteinuria (r=0.37; P=0.002)
- Considering all visits (CRR present/absent=146/40), mean ± SD RAIL-scores of patients with CRR-status were 1.17 ± 1.53 lower (P=0.023; area under the ROC curve=0.73) than those without CRR
- RAIL scores of no more than 3.4 identified CRR-status with 78% specificity (positive predictive value=0.77, sensitivity=62; **Figure 1**)
- Patients who newly achieved CRR status at the next visit had a RAIL-score mean ± SD decrease of 1.0 ± 1.524 since the last visit (P=0.0024), and patients with a RAIL-score decrease of >0.57 had a percentage variance value of 82% of achieving CRR at the next visit

TABLE 1: Patient characteristics & RAIL-scores over time

	Enrollment (T0) (n=69)	6 months (T1) (n=58)	12 months (T2) (n=59)
Gender, Female / Male, n (%)	63 (91) / 6 (9)		
Ethnicity, Caucasian / Mestizo, n (%)	10 (15.5) / 58 (84)		
Age at entry, years, median (IQR)	37 (25, 44)		
Time to diagnosis, months, median (IQR)	28 (21, 36)		
LN, Yes / No, n (%)	51 (74) / 18 (26)		
Complete Renal Response, n (%)	37 (54)	51 (88)	55 (93)
RAIL-Cr score, median (IQR)	3.74 (2.72, 4.88)	3.47 (2.47, 4.12)	3.46 (2.43, 4.62)
Proteinuria, median (IQR)	0.36 (0.12, 1.7)	0.16 (0.1, 0.3)	0.13 (0.1, 0.29)
Hematuria, n (%)	19 (28)	7 (12)	3 (5)
C3 / C4, low, n (%)	61 (88) / 57 (83)	14 (24) / 15 (26)	16 (27) / 18 (31)
SLEDAI total score, median (IQR)	6 (2, 13)	2 (0, 4)	2 (0, 4)
SDI total score, median (IQR)	0 (0, 1)	0 (0, 1)	0 (0, 1)
Treatments, n (%)			
ACE/ARB	37 (54)	28 (48)	23 (39)
Azathioprine	9 (13)	5 (9)	10 (17)
Prednisone / Pulsed Steroids	56 (81) / 12 (17)	44 (76) / 7 (12)	41 (69) / 3 (5)
Antimalarial	65 (94)	51 (88)	58 (98)
Cyclophosphamide	9 (13)	12 (21)	4 (7)
Mycophenolate mofetil	24 (35)	21 (36)	34 (58)
Tacrolimus	3 (4)	2 (3.5)	4 (7)

__ACE__=Angiotensin-converting enzyme, __ARB__=Angiotensin receptor blockers, __Cr__=creatinine, __IQR__=Interquartile range, __LN__=Lupus nephritis, __RAIL__=Renal Activity Index for Lupus, __SDI__=SLICC/ACR Damage Index, __SLE__=Systemic lupus erythematosus, __SLEDAI__=Systemic Lupus Erythematosus Disease Activity Index.

CONCLUSIONS

In patients with SLE, CRR status is associated with lower RAIL scores; RAIL score decreases of >0.57 between follow-up visits are predictive of future CRR achievement

FIGURE 1: ROC Curve for Model

Area Under the Curve=0.7264

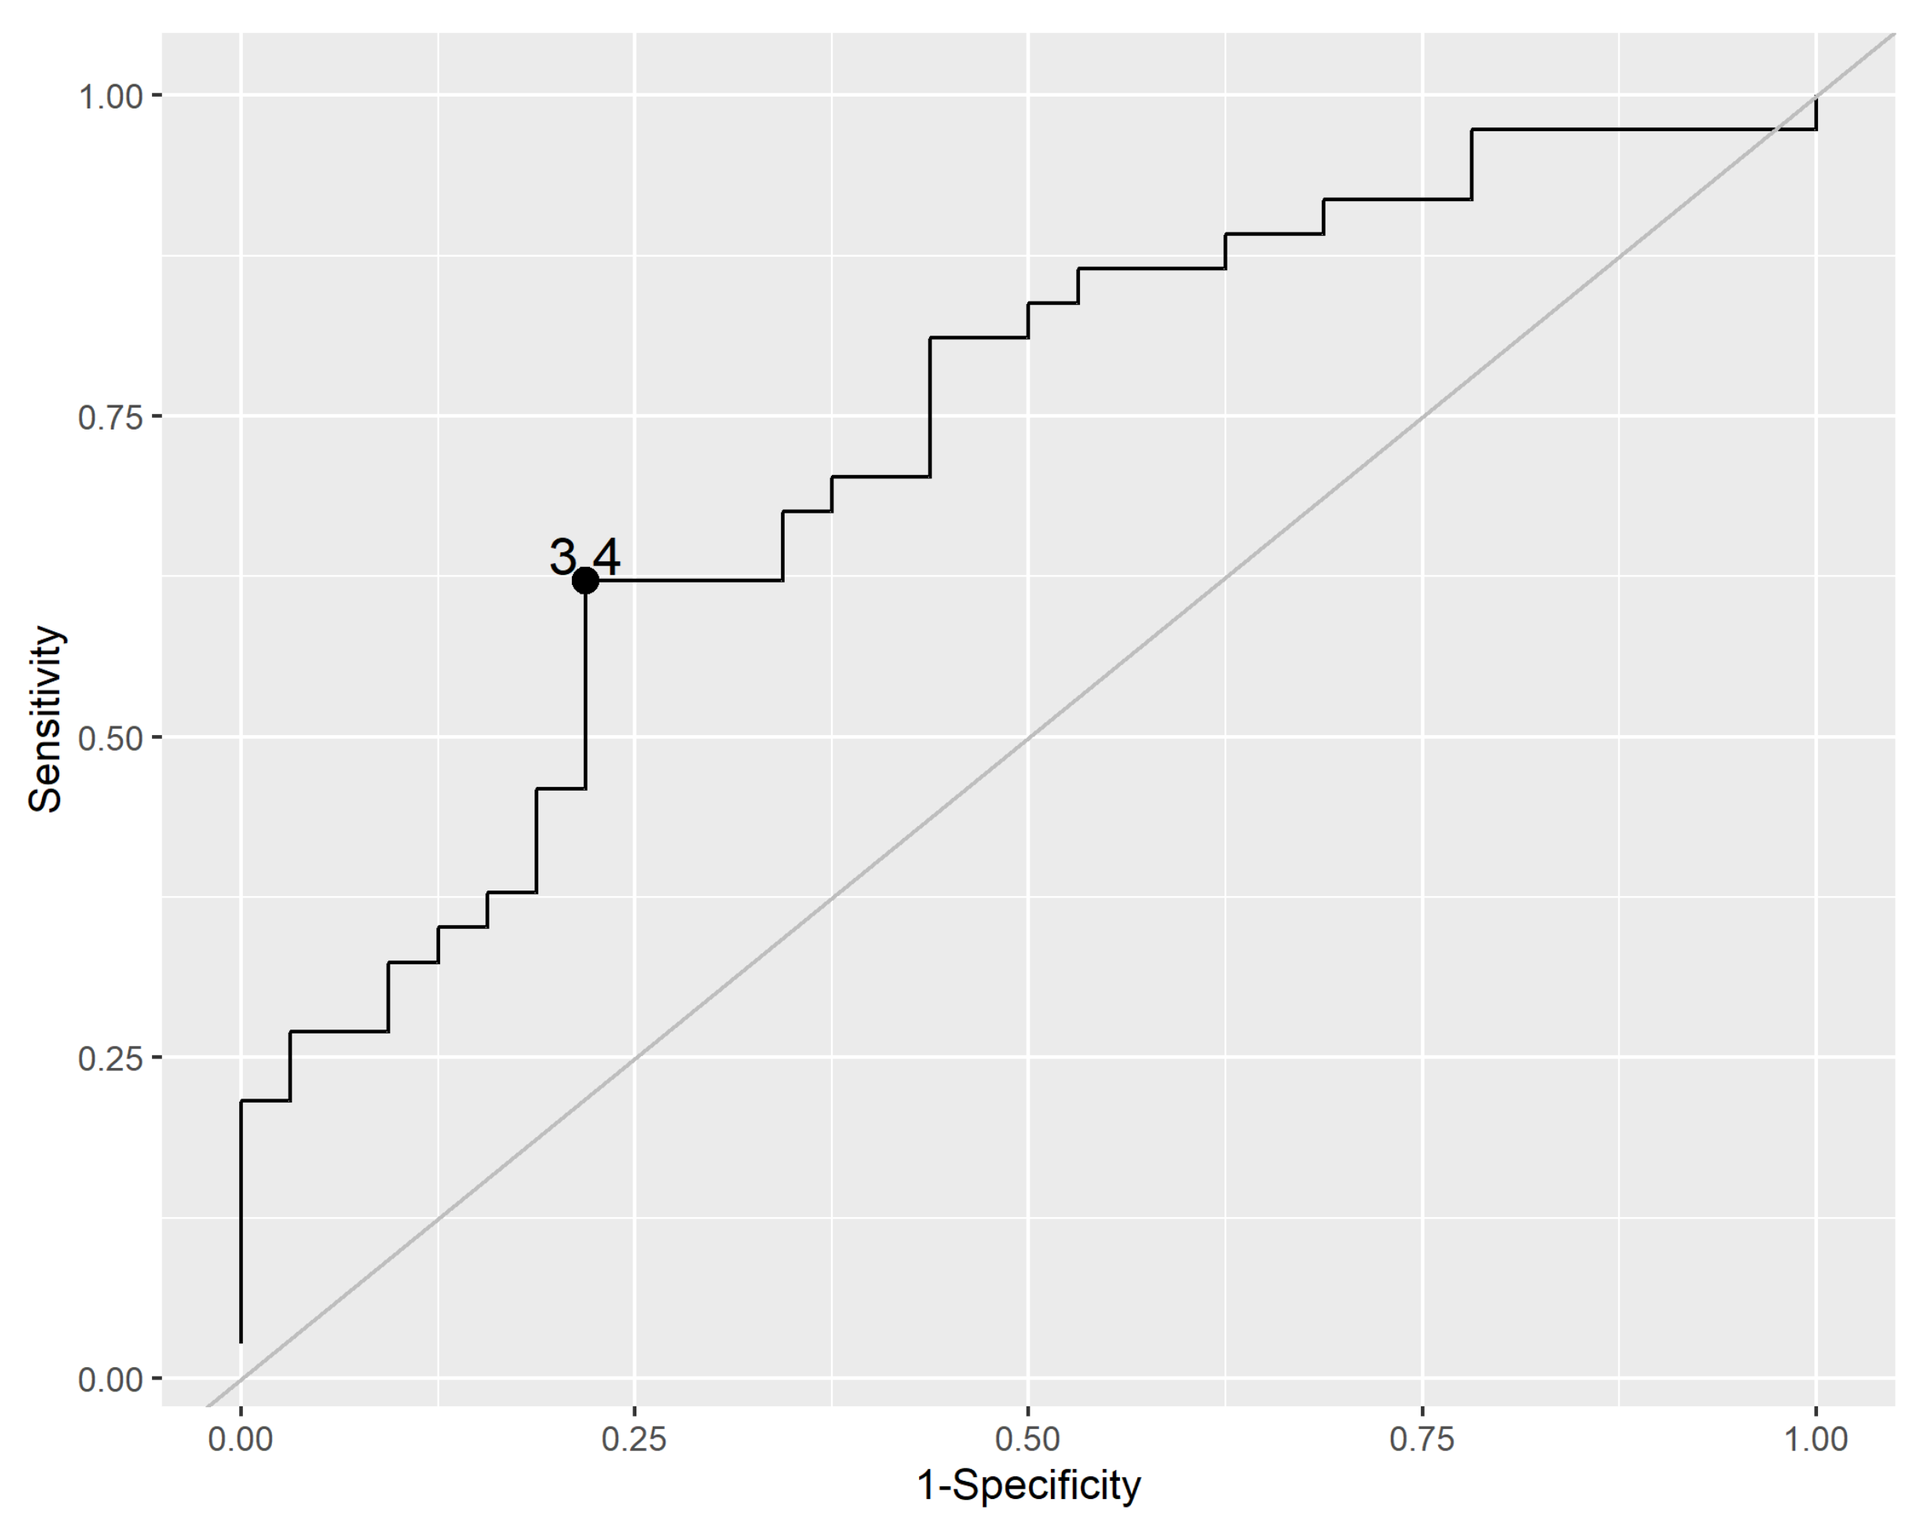
Sensitivity
1-Specificity
3.4
0.00
0.25
0.50
0.75
1.00

REFERENCES

1. Brunner HI, et al. *Arthritis Care Res (Hoboken)*. 2016;68(7):1003–11.

ACKNOWLEDGMENTS

This study was sponsored by Johnson & Johnson. Medical writing support was provided by Namiko Abe, PhD, of Joulé Clinical, and was funded by Johnson & Johnson. Layout design and reformatting for this encore presentation was provided by Samita Warang of Siro Clinpharm Pvt. Ltd., India (funded by Johnson & Johnson).

DISCLOSURES

G. Pons-Estel: Served as a speaker, advisor, and/or on steering committees for AstraZeneca, Boehringer Ingelheim, GSK, Janssen, Novartis, Pfizer, RemeGen, Sanofi, and Werfen Diagnostics; and has received grants and consulting fees from these same companies. **R. Quintana, R. Nieto, C. Funes Soaje, P. Alba, V. Saurit, M. A. García, G. A. Berbotto, V. Bellomio, E. M. Kerzberg, G. N. Gómez, C. Pisoni, V. Juarez, A. Malvar, N. A. da Silva, O. A. Monticielo, H. A. Mariz, F. M. Ribeiro, E. F. Borba, E. Bonfa, E. T. dos Reis Neto, I. G. Herrera, L. Massardo, G. Aroca Martinez, L. G. Escorcia, C. A. Cañas Davila, G. Quintana-Lopez, C. E. Toro-Gutierrez, M.M. Alvarez, M.A. Saavedra Salinas, M. P. Hernandez, H. Fragoso-Loyo, L. H. Silveira, I. García-De La Torre, C. Abud Mendoza, J. A. Esquivel-Valerio, I. Acosta Colman, A. Paats, C. Mora Trujillo, A. C. Quiroz, R. M. Louis, M. Rebella,** and **Á. Danza:** Declare no conflicts of interest. **H. Brunner:** Received support from AstraZeneca, Boehringer Ingelheim, Novartis, Pfizer, UCB, BMS, Takeda. **M. Scolnik:** Received speaker fees and/or served in advisory roles for GSK, AstraZeneca, Janssen, Roche, and Pfizer; and has received grants from GSK, AstraZeneca, Janssen, Roche, and Pfizer. **M. F. Ugarte-Gil:** Received grant support from Janssen; received consulting and/or speaking fees from AstraZeneca, Ferrer, GSK, Novartis and Tecnofarma. **F. Zazzetti, A. Orillion:** Employees of Johnson & Johnson and may hold stocks or stock options in Johnson & Johnson. **B. Pons-Estel:** Served as a speaker and/or advisor for AstraZeneca, GSK, and Janssen. Previously presented at European Alliance of Associations for Rheumatology (Eular); Barcelona, Spain; June 11–14, 2025

PRESENTED AT: AMERICAN COLLEGE OF RHEUMATOLOGY (ACR) CONVERGENCE 2025; CHICAGO, IL, USA; OCTOBER 24–29, 2025.