1486

SYSTEMIC LUPUS ERYTHEMATOSUS AND STATINS IN GLADEL 2.0: ARE CARDIOVASCULAR RISK PREVENTION GUIDELINES BEING FOLLOWED?

ACR Convergence Where Rheumatology Meets

GLADEL LATIN AMERICA

Ileana Cecilia Reynosa-Silva*, Iris Jazmin Colunga-Pedraza, Guillermina Harvey, Dionicio Ángel Galarza-Delgado, José Ramón Azpiri-López, Fernanda Massiel García García, Oscar Azael Garza Flores, Marina Scolnik, Gisela Subils, Cintia Otaduy, Verónica Saurit, Valeria Arturi, Leonel Berbotto, Guillermo Pons-Estel, Luciana Gonzalez Lucero, Melissa Brenda Serna Góngora, María de los Angéles Gargiulo, Cecilia Nora Pisoni, Maria Elena Crespo, Mónica Sánchez Guamán, Vitalina de Souza Barbosa, Andrese Aline Gasparin, Henrique Ataide Mariz, Francinne Machado Ribeiro, Eduardo F. Borba, Edgard Torres dos Reis-Neto, Iris Guerra Herrera, Loreto Massardo, Gustavo Aroca Martínez, Carlos A. Cañas, Gerardo Quintana-Lopez, Carlos Enrique Toro Gutiérrez, Mario Javier Moreno Alvarez, Miguel Ángel Saavedra, Margarita Portela Hernández, Hilda Fragoso-Loyo, Luis M. Amezcua-Guerra, Ignacio García de la Torre, Jorge Isaac Velasco Santos, Jorge A. Esquivel Valerio, Jhonatan Losanto, Magaly Alva Linares, Manuel F. Ugarte-Gil, Katiuzka Zuñiga Corrales, Roberto Muñoz Louis, Carina Pizzarossa, Gonzalo Silveira, Federico Zazzetti, Ashley Orillion, Gloria Vásquez, on behalf of Grupo Latino Americano De Estudio de Lupus (GLADEL), Study Group of PANLAR

*Presenting author

BACKGROUND

- Patients with systemic lupus erythematosus (SLE) have a significantly increased risk of atherosclerotic cardiovascular disease (ASCVD)
- European Alliance of Associations for Rheumatology (EULAR) recommends a comprehensive assessment and management of cardiovascular risk (CVR), following general population guidelines[1]
- Statin therapy plays a key role in reducing CVR and preventing ASCVD events[2]

OBJECTIVE

- This study aimed to determine the proportion of patients with SLE eligible for statin use for primary ASCVD prevention, based on American and European CVR guidelines

METHODS

- This was an observational and cross-sectional study of GLADEL 2.0, a multi-ethnic Latin-American SLE cohort
 - Demographics, comorbidities, medications, disease activity, and laboratory data were analyzed
- Statin eligibility was determined using the 2019 American College of Cardiology/American Heart Association (ACC/AHA) and the 2021 European Society of Cardiology (ESC) guidelines
- CVR was assessed using the ASCVD risk calculator from the ACC/AHA, Systemic Coronary Risk Evaluation 2 (SCORE2), and Pan American Health Organization (PAHO) risk scores
- Cohen's Kappa coefficient was used to determine inter-guideline agreement
- Both guidelines only consider paients ≥40 years of age as candidates for CVR prevention with statins, due to eligibility dependence on CVR scores
 - Therefore, a comparative analysis was conducted between patients above and below this age threshold

RESULTS

- Among 1083 patients in the GLADEL 2.0 SLE cohort, only 394 (36.4%) were older than 40 years of age (**Table 1**)
- ACC/AHA could only be calculated for 164 patients, PAHO for 351 patients, and SCORE2 for 181 patients
- Most of these patients were categorized as having a low CVR, regardless of the calculator used
- A total of 15 patients were indicated to receive statin therapy based on European guidelines; among these, only 5 (33%) had a previous prescription for a statin
- Among 50 patients eligible for statin therapy based on American guidelines, only 13 (26%) had been prescribed them
- Inter-guideline agreement on statin eligibility was fair (Cohen's Kappa = 0.35; 95% CI: 0.15–0.55)
- Patients under 40 years of age were more obese and had higher disease activity compared with those older than 40 years of age (**Table 2**)

CONCLUSIONS

- This study reveals a gap in the management of CVR among patients with SLE in the GLADEL 2.0 cohort, as only a small percentage are candidates for statin therapy, primarily due to the predominance of younger patients under age 40
 - Traditional CVR assessment tools fail to encompass this group, which is at an elevated risk for accelerated atherosclerosis
- Among eligible patients, statin prescribing rates remain low, suggesting a missed opportunity in proactive cardiovascular prevention
- The moderate agreement between differing guidelines highlights the inconsistency in risk assessment and management approaches for this population
- There is a need for tailored strategies to comprehensively evaluate and address CVR in younger patients with chronic inflammatory conditions like SLE
- Implementing more inclusive guidelines could enhance preventative measures and improve outcomes for patients at increased risk for ASCVD

TABLE 1.
Baseline clinical characteristics, disease activity, damage index and treatments

	TOTAL (N=1083)		TOTAL (N=1083)
Age, years, median (Q1, Q3)	35.4 (15.6, 77.5)	**Triglycerides (mg/dL)**, median (Q1, Q3), n=557	124 (32, 619)
≥40 years, n (%)	394 (36.4)	**Diabetes**, n (%)	45 (4.2)
Female, n (%)	970 (89.6)	**Hypertension**, n (%)	293 (27.2)
Mestizo, n (%)	701 (64.7)	**Dyslipidemia**, n (%)	129 (12)
Obesity, n (%)	187 (18.5)	**Active smoking**, n (%)	54 (5.1)
Disease duration, years, median (Q1, Q3)	5.6 (0.6, 61.6)	**Chronic kidney disease**, n (%)	61 (5.6)
SLEDAI, median (Q1, Q3)	5 (0, 57)	**Acute myocardial infarction**, n (%)	8 (0.7)
SDI, median (Q1, Q3)	0 (0, 9)	**Statins**, n (%)	155 (20.4)
HDL-c (mg/dL), median (Q1, Q3), n=504	49 (11, 102)	**Prednisone**, n (%)	758 (72.5)
LDL-c (mg/dL), median (Q1, Q3), n=480	103 (28, 328)	**Hydroxychloroquine**, n (%)	876 (84)

__HDL-c__=high-density lipoprotein cholesterol, __LDL-c__=low-density lipoprotein cholesterol, __SLEDAI__=Systemic Lupus Erythematosus Disease Activity Index, __SDI__=Systemic Lupus International Collaborating Clinics/American College of Rheumatology Damage Index.

TABLE 2.
Comparison of clinical characteristics based on their age group

	<40 YEARS	≥40 YEARS	*P* VALUE
Obesity, n (%)	102 (15.8)	85 (23.3)	0.0031[a]
Disease duration, years, median (Q1, Q3)	4.1 (0.2 28.9)	9.9 (0.6, 61.6)	<0.0001[b]
SLEDAI, median (Q1, Q3)	6 (0, 57)	3 (0, 38)	<0.0001[b]
HDL-c, mg/dL, median (Q1, Q3), n=504	47 (11, 102)	52 (15, 100)	0.0003[b]
LDL-c, mg/dL, median (Q1, Q3), n=480	105.5 (37, 241)	101 (28, 328)	0.1404[b]
Triglycerides, mg/dL, median (Q1, Q3), n=557	123 (39, 619)	127 (32, 525)	0.4044[b]
Diabetes, n (%)	12 (1.7)	33 (8.4)	<0.0001[a]
Active smoking, n (%)	30 (4.5)	24 (6.2)	0.0503[a]
Statins, n (%)	94 (19.8)	61 (21.3)	0.0510[a]
Prednisone, n (%)	525 (78.5)	233 (61.8)	<0.0001[a]

[a]Chi-Square p-value. [b]Kruskal-Wallis p-value. __HDL-c__=high-density lipoprotein cholesterol, __LDL-c__=low-density lipoprotein cholesterol, __SLEDAI__=Systemic Lupus Erythematosus Disease Activity Index.

REFERENCES

1. Drosos GC, et al. *Ann Rheum Dis.* 2022;81(6):768–779.
2. Yousef Yengej FA, et al. *Neth J Med.* 75(3):99–105.

ACKNOWLEDGMENTS

This study was sponsored by Johnson & Johnson. Medical writing support was provided by Kaushik Kuche, PhD; poster layout and graphic design support were provided by Amit Kavle, both of SIRO Medical Writing Pvt. Ltd., India (funded by Johnson & Johnson).

DISCLOSURES

MS: Received speaker fees, advisory board fees, and/or grants from GSK, AstraZeneca, Janssen, Roche, and Pfizer. **GP-E:** Received grants and consulting fees from and participated as a speaker, as an advisor, and/or on a steering committee for AstraZeneca, Boehringer Ingelheim, GSK, Janssen, Novartis, Pfizer, RemeGen, Sanofi, and Werfen Diagnostics. **MFU-G:** Received grant support from Janssen; received consulting and/or speaking fees from AstraZeneca, Ferrer, GSK, Novartis and Tecnofarma. **FZ, AO:** Employment—Johnson & Johnson, stock ownership—Johnson & Johnson. **ICR-S, IJC-P, GH, DAG-D, JRA-L, FMGG, OAGF, GS, CO, VS, VA, LB, LGL, MBSG, MdlAG, CNP, MEC, MSG, VdSB, AAG, HAM, FMR, EFB, ETdR-N, IGH, LM, GAM, CAC, GQ-L, CETG, MJMA, MAS, MPH, HF-L, LMA-G, IGdlT, JIVS, JAEV, JL, MAL, KZC, RML, CP, GS, GV:** Declared no conflicts of interest.

PRESENTED AT: AMERICAN COLLEGE OF RHEUMATOLOGY (ACR) CONVERGENCE OCTOBER 24–29, 2025; CHICAGO, IL, USA.