1491

FEASIBILITY OF EXTRARENAL SYSTEMIC LUPUS ERYTHEMATOSUS DISEASE MODIFICATION IN GLADEL 2.0, A LATIN AMERICAN COHORT

ACR Convergence Where Rheumatology Meets

GLADEL LATIN AMERICA

Julián E. Barahona-Correa*, Santiago Bernal-Macías, Daniel Fernández-Ávila, Óscar Muñoz, Lucia Hernández, Erika Susana Palacios Santillan, Laura Maurelli, Paula Alba, Verónica Saurit, Lucila García, María Emilia Sattler, Maria Constanza Bertolaccini, Marina Laura Micelli, Graciela Noemí Gómez, Micaela A. Cosatti, Ana Carolina Ralle, Joaquín Martinez Serventi, Ana Carolina O. S. Montandon, Odirlei Andre Monticielo, Ángela Luzia Branco Pinto Duarte, Laíssa Cristina Alves Alvino, Eduardo F. Borba, Eloisa Bonfa, Edgard Torres dos Reis-Neto, Iris Guerra Herrera, Milena Mimica, Gustavo Aroca Martínez, Lorena Gómez Escorcia, Carlos A. Cañas, Gerardo Quintana-Lopez, Carlos Enrique Toro Gutiérrez, José Maximiliano Martínez Pérez, Reyna Elizabeth Sánchez-Briones, Mario Pérez Cristóbal, Eduardo Martin-Nares, Yaneli Juárez-Vicuña, Ignacio García-Valladares, Rodrigo Ortiz Hernández, Jorge A. Esquivel Valerio, Isabel Acosta Colman, Astrid Paats, Jorge Cieza Calderón, Manuel F. Ugarte-Gil, Armando Calvo Quiroz, Rodamin Alvarez, Analía Cánepa, Carina Pizzarossa, Federico Zazzetti, Ashley Orillion, Cristina Drenkard, on behalf of Grupo Latino Americano De Estudio de Lupus (GLADEL), Study Group of PANLAR

*Presenting author

BACKGROUND

- The concept of disease modification (DM) has been recently introduced to improve the long-term care of patients with systemic lupus erythematosus (SLE)
- DM is defined as 'the minimization of disease activity with the fewest treatment-associated toxicities and slowing/preventing organ damage progression'.
 - To claim DM ultimately requires delayed or prevented progression of organ damage beyond 5 years
- The concept of DM is valuable for assessing 'whether an intervention is on track for achieving DM at the 5-year mark,' based on interim evaluations during the first 5 years
- Latin American patients with SLE are prone to poorer outcomes, which are associated with both disease severity and social determinants of health (SDH)

OBJECTIVE

- This preliminary study evaluates the prevalence of extrarenal DM in Latin American patients with SLE and examines differences in SDH and treatment between those who did and did not achieve extrarenal DM

METHODS

- GLADEL 2.0 is a longitudinal cohort assessing the incidence and prevalence of SLE
- Forty-three centers from 10 Latin-American countries enrolled patients ≥18 years of age who fulfilled the 1982/1997 ACR and/or the 2012 Systemic Lupus International Collaborating Clinics (SLICC) classification criteria for SLE
- The composite definition of DM for extrarenal SLE included:
 - (i) a significant reduction in SLE Disease Activity Index (SLEDAI; >3 points),
 - (ii) no severe flares, and
 - (iii) ≤10 mg/day of prednisone for months 0–12 and ≤5 mg/day prednisone-equivalent for years 2–5
- Patients with complete data at baseline, 12, 24, 36, 48, and 60 months were analyzed
- We compared baseline SDH and exposure to medications between patients who did and did not achieve DM criteria over time

RESULTS

- Among 1,083 patients who entered the GLADEL 2.0 cohort, 709 had the baseline data needed to fulfill DM criteria. **Table 1** shows differences in demographic and SDH data by extrarenal DM achievement groups over time
 - Data from month 60 were not analyzed as data were available for only 64 patients
- Although educational attainment varied between groups and genders at each time point, a higher proportion of patients achieved DM among men compared to women at the 36 and 48-month marks
- Overall, the proportion of patients achieving DM criteria ranged between 16–20% across time points (**Figure 1A**)
- **Figure 1B** shows DM trajectories during follow-up
- Medication exposure is depicted in **Figure 2**
 - A higher number of patients achieving extrarenal DM at months 36 and 48 received cyclophosphamide

TABLE 1.
Sociodemographics and their differences among patients with SLE who did and did not achieve extrarenal disease modification over time

	Extrarenal Disease Modification at Each Time Point							
	Month 12 (N=709)		***Month 24 (N=663)***		***Month 36 (N=460)***		***Month 48 (N=379)***	
Baseline Descriptor	**Yes (N=111)**	**No (N=598)**	**Yes (N=115)**	**No (N=548)**	**Yes (N=92)**	**No (N=368)**	**Yes (N=73)**	**No (N=306)**
Age at diagnosis, median (IQR)	26 (19–33.5)	27 (20–36)	27 (20–36)	26 (20–34)	27 (21–37)	26 (20–33)	29 (21–38)	27 (20–35)
Age at enrollment, median (IQR)	33 (25.5–42)	36 (27–45)	33 (27–42)	36 (28–45)	34.5 (27.8–44)	36 (28–46)	35 (29–45)	38 (29–47)
Sex at birth, n (%)								
Female	104 (93.7%)	534 (89.3%)	101 (87.8%)	496 (90.5%)	**75 (81.5%)**	**339 (92.1%)**	**59 (80.8%)**	**280 (91.5%)**
Male	7 (6.3%)	64 (10.7%)	14 (12.2%)	52 (9.5%)	**17 (18.5%)**	**29 (7.9%)**	**14 (19.2%)**	**26 (8.5%)**
Ethnicity, n(%)								
White	30 (27.3%)	183 (30.8%)	37 (32.5%)	153 (28.1%)	24 (26)	108 (29.7%)	23 (31.5%)	94 (31.1%)
Mixed	68 (61.8%)	363 (61.1%)	71 (62.3%)	344 (63.2%)	56 (61.5%)	228 (62.6%)	43 (58.9%)	183 (60.6%)
African Latin American	12 (10.9%)	47 (7.9%)	6 (5.3%)	47 (8.6%)	11 (12.1%)	28 (7.7%)	7 (9.6%)	25 (8.3%)
Other		1 (0.2%)						
Country of birth, n (%)								
Argentina	35 (31.5%)	193 (32.3%)	36 (31.3%)	151 (27.6%)	30 (32.6%)	107 (29.1%)	21 (28.8%)	89 (29.1%)
Brazil	21 (18.9%)	128 (21.4%)	26 (22.6%)	125 (22.8%)	29 (31.5%)	72 (19.6%)	20 (27.4%)	64 (20.9%)
Chile	2 (1.8%)	23 (3.8%)	2 (1.7%)	22 (4%)	-	14 (3.8%)	-	5 (1.6%)
Colombia	18 (16.2%)	40 (6.7%)	6 (5.2%)	31 (5.7%)	2 (2.2%)	16 (4.3%)	4 (5.5%)	21 (6.9%)
Dominican Republic	4 (3.6%)	15 (2.5%)	2 (1.7%)	12 (2.2%)	3 (3.3%)	9 (2.4%)	2 (2.7%)	8 (2.6%)
Ecuador	2 (1.8%)	12 (2%)	1 (0.9%)	8 (1.5%)	-	8 (2.2%)	-	4 (1.3%)
Honduras	-	1 (0.2%)	-	-	-	-	-	-
Mexico	15 (13.5%)	81 (13.5%)	15 (13%)	88 (16.1%)	15 (16.3%)	55 (14.9%)	19 (26%)	52 (17%)
Paraguay	4 (3.6%)	43 (7.2%)	8 (7%)	34 (6.2%)	7 (7.6%)	26 (7.1%)	1 (1.4%)	16 (5.2%)
Peru	7 (6.3%)	27 (4.5%)	11 (9.6%)	44 (8%)	3 (3.3%)	34 (9.2%)	3 (4.1%)	30 (9.8%)
Uruguay	3 (2.7%)	33 (5.5%)	7 (6.1%)	32 (5.8%)	3 (3.3%)	27 (7.3%)	3 (4.1%)	17 (5.6%)
Venezuela	-	2 (0.3%)	1 (0.9%)	1 (0.2%)	-	-	-	-
Marital status, n (%)								
Married-Concubine	45 (40.5%)	247 (43%)	55 (48.7%)	233 (43.3%)	41 (45.6%)	154 (42.7%)	31 (44.9%)	133 (44.9%)
Separated-Divorced	4 (3.6%)	38 (6.6%)	7 (6.2%)	37 (6.9%)	7 (7.8%)	28 (7.8%)	6 (8.7%)	20 (6.8%)
Single	60 (54.1%)	281 (49%)	49 (43.4%)	260 (48.3%)	40 (44.4%)	175 (48.5%)	30 (43.5%)	139 (47%)
Widow	2 (1.8%)	8 (1.4%)	2 (1.8%)	8 (1.5%)	2 (2.2%)	4 (1.1%)	2 (2.9%)	4 (1.4%)
Education (years), median (IQR)	**14 (11–16)**	**13 (11–16)**	**14 (11–16)**	**13 (11–16)**	**12.5 (11–16)**	**13 (11–16)**	**12 (11–16)**	**13 (11–16)**
Employment status, n (%)								
Full/Partial Work	**52 (49.5%)**	**322 (56.9%)**	61 (55%)	283 (54.7%)	41 (46.1%)	207 (58%)	38 (53.5%)	165 (55.7%)
Retired	**2 (1.9%)**	**33 (5.8%)**	3 (2.7%)	23 (4.4%)	4 (4.5%)	20 (5.6%)	6 (8.5%)	19 (6.4%)
Student	**18 (17.1%)**	**55 (9.7%)**	13 (11.7%)	52 (10.1%)	12 (13.5%)	29 (8.1%)	4 (5.6%)	24 (8.1%)
Unemployed	**33 (31.4%)**	**156 (27.6%)**	34 (30.6%)	159 (30.8%)	32 (36%)	101 (28.3%)	23 (32.4%)	88 (29.7%)
Socioeconomic status, n (%)								
High	2 (1.9%)	16 (2.7%)	4 (3.5%)	13 (2.4%)	**5 (5.5%)**	**9 (2.4%)**	4 (5.6%)	11 (3.6%)
Middle high	18 (16.7%)	131 (22.3%)	21 (18.6%)	119 (22.1%)	**11 (12.1%)**	**87 (23.6%)**	10 (14.1%)	64 (20.9%)
Middle	43 (39.8%)	200 (34%)	48 (42.5%)	178 (33.1%)	**31 (34.1%)**	**130 (35.3%)**	27 (38%)	124 (40.5%)
Middle low	31 (28.7%)	175 (29.8%)	24 (21.2%)	170 (31.6%)	**28 (30.8%)**	**107 (29.1%)**	20 (28.2%)	84 (27.5%)
Low	14 (13%)	66 (11.2%)	16 (14.2%)	58 (10.8%)	**16 (17.6%)**	**35 (9.5%)**	10 (14.1%)	23 (7.5%)
Medical coverage, n (%)								
Full coverage	66 (60%)	299 (50.7%)	64 (57.7%)	297 (54.6%)	**44 (49.4%)**	**215 (59.1%)**	39 (54.9%)	186 (61.4%)
Partial coverage	14 (12.7%)	85 (14.4%)	11 (9.9%)	75 (13.8%)	**8 (9%)**	**56 (15.4%)**	13 (18.3%)	45 (14.9%)
Without coverage	30 (27.3%)	206 (34.9%)	36 (32.4%)	172 (31.6%)	**37 (41.6%)**	**93 (25.5%)**	19 (26.8%)	72 (23.8%)

*Composite definition of extrarenal SLE disease modification included: (i) a significant reduction in SLEDAI (>3 points), (ii) no severe flares , and (iii) ≤10 mg/day of prednisone for months 0–12 and ≤5 mg/day prednisone-equivalent for years 2–5. Values presented in bold represent significant differences of baseline descriptors between disease modification groups at the corresponding time point ($p<0.05$). The number of patients decreased at each time point due to either insufficient time in the cohort or loss of follow-up. Comparisons were performed using a Chi-squared test for qualitative variables or a non-parametric t-test for continuous variables. **IQR**=interquartile range, **SLE**=systemic lupus erythematosus, **SLEDAI**=SLE Disease Activity Index.*

CONCLUSIONS

- In this preliminary investigation, up to 20% of Latin American patients with SLE achieved extrarenal DM
- The study highlights critical disparities, indicating that patients with lower educational attainment and women were less likely to achieve DM
- Our findings suggest that specific medication exposure may play a significant role in extrarenal DM achievement; further research may focus on the impact of cyclophosphamide given a higher percentage of patients achieving DM in that group
- These results underscore the importance of addressing SDH and tailoring treatment strategies to improve outcomes for marginalized groups within this population
- Continued research is essential to discern the independent contributions of SDH and different treatment modalities on achieving extrarenal DM, ultimately aiming to enhance long-term care and patient quality of life

FIGURE 1.
Percentage of patients achieving potential for extrarenal SLE disease modification (A) and disease modification trajectory (B)

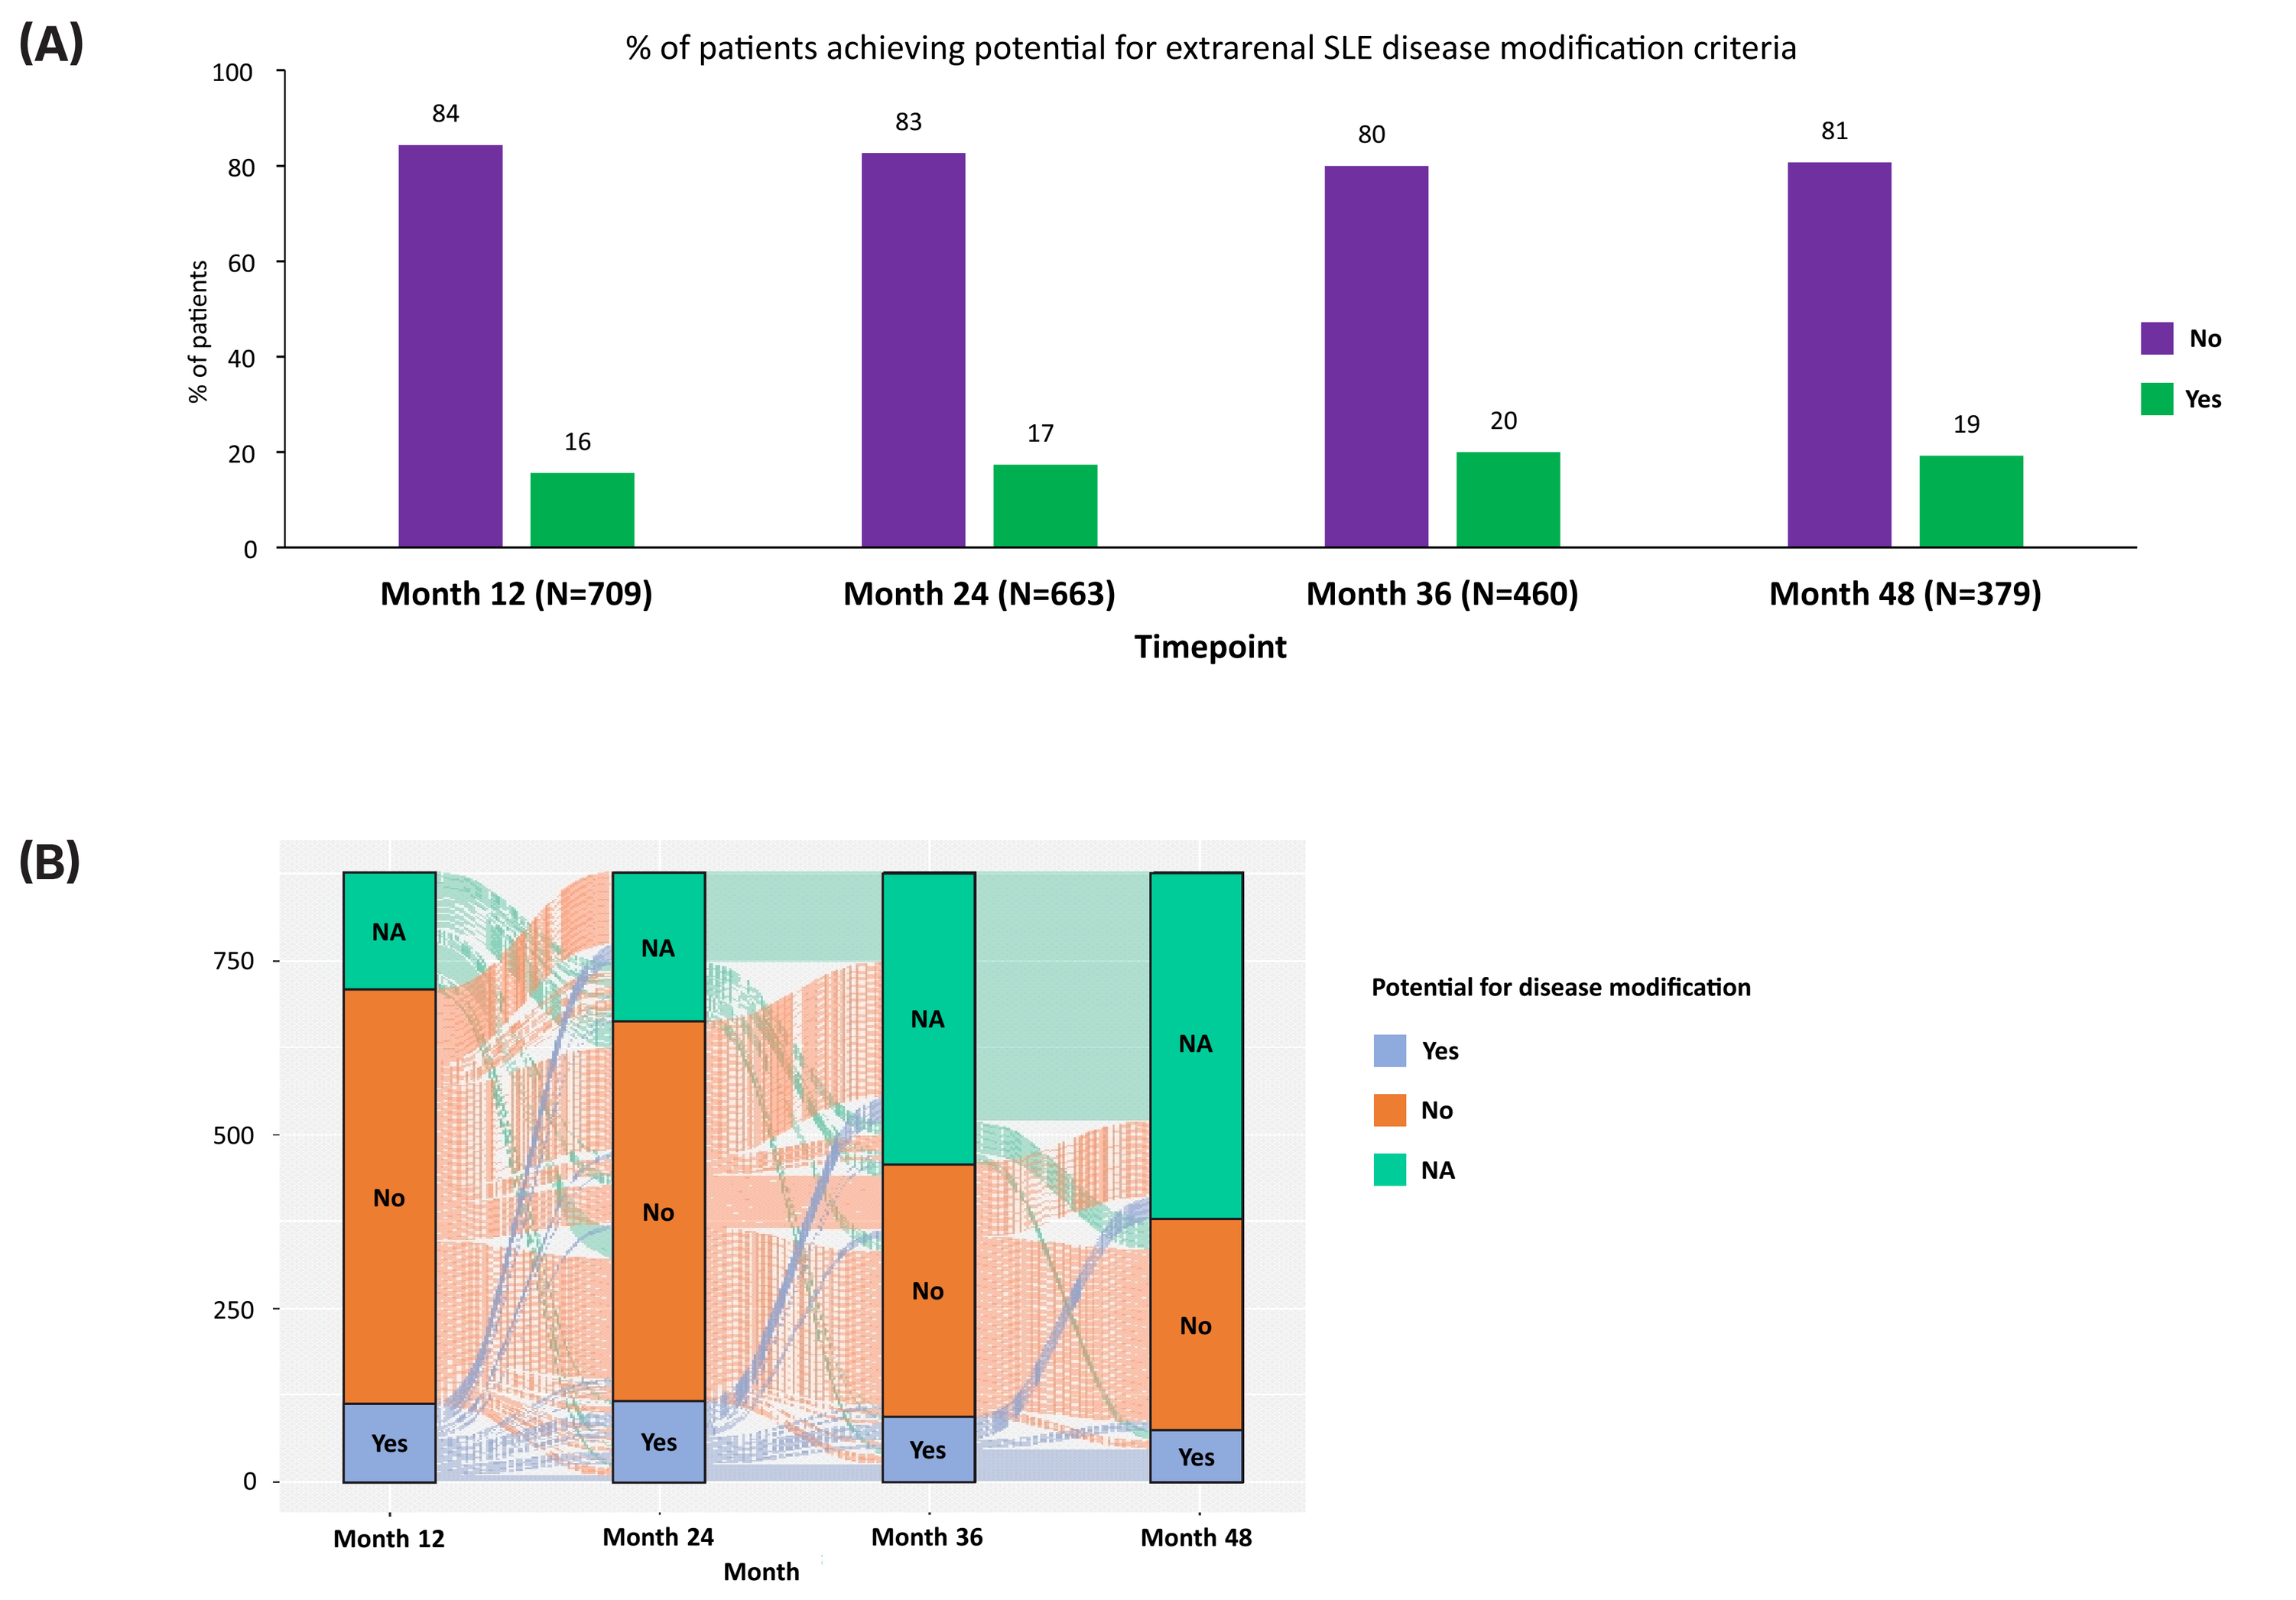
(A)
% of patients achieving potential for extrarenal SLE disease modification criteria
% of patients
84
16
83
17
80
20
81
19
No
Yes
Month 12 (N=709)
Month 24 (N=663)
Month 36 (N=460)
Month 48 (N=379)
Timepoint
(B)
Potential for disease modification
Yes
No
NA
Month 12
Month 24
Month 36
Month 48
Month

FIGURE 2.
Medication exposure in patients who did (A) and (B) did not achieve DM

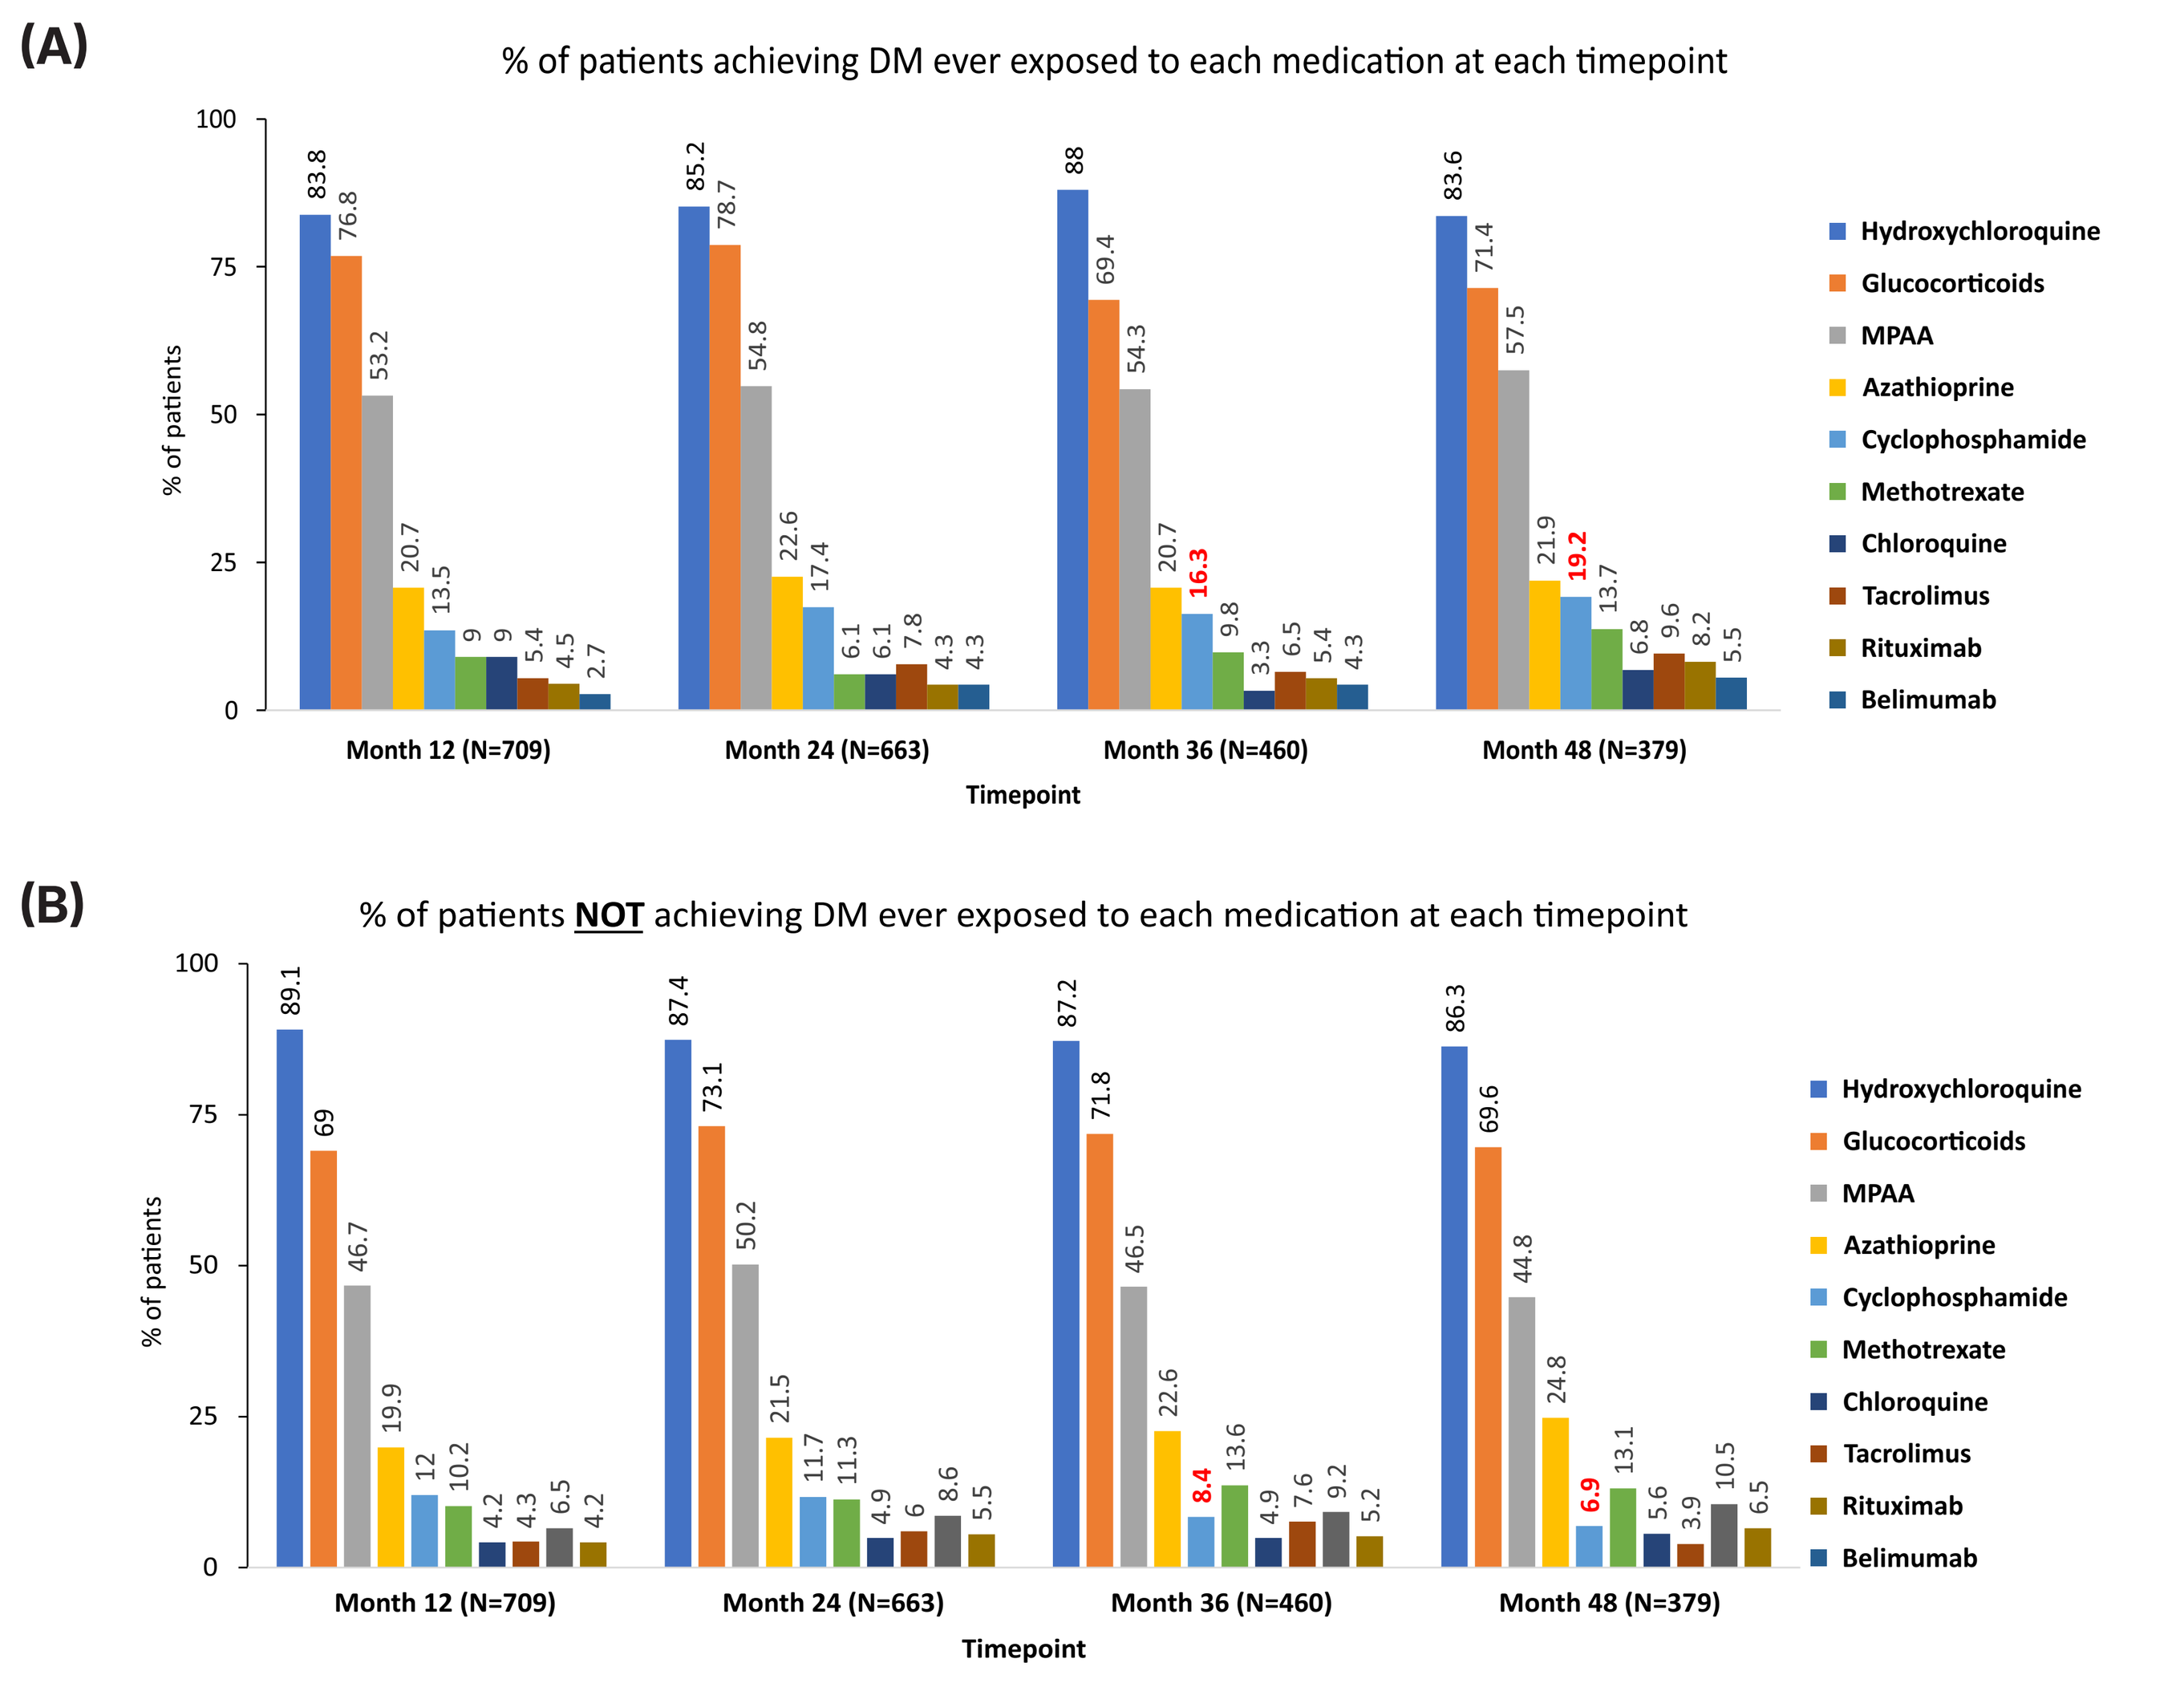
(A)
% of patients achieving DM ever exposed to each medication at each timepoint
% of patients
Month 12 (N=709): 83.8, 76.8, 53.2, 20.7, 13.5, 9, 9, 5.4, 4.5, 2.7
Month 24 (N=663): 85.2, 78.7, 54.8, 22.6, 17.4, 6.1, 6.1, 7.8, 4.3, 4.3
Month 36 (N=460): 88, 69.4, 54.3, 20.7, 16.3, 9.8, 3.3, 6.5, 5.4, 4.3
Month 48 (N=379): 83.6, 71.4, 57.5, 21.9, 19.2, 13.7, 6.8, 9.6, 8.2, 5.5
Timepoint
Hydroxychloroquine
Glucocorticoids
MPAA
Azathioprine
Cyclophosphamide
Methotrexate
Chloroquine
Tacrolimus
Rituximab
Belimumab
(B)
% of patients NOT achieving DM ever exposed to each medication at each timepoint
% of patients
Month 12 (N=709): 89.1, 69, 46.7, 19.9, 12, 10.2, 4.2, 4.3, 6.5, 4.2
Month 24 (N=663): 87.4, 73.1, 50.2, 21.5, 11.7, 11.3, 4.9, 6, 8.6, 5.5
Month 36 (N=460): 87.2, 71.8, 46.5, 22.6, 8.4, 13.6, 4.9, 7.6, 9.2, 5.2
Month 48 (N=379): 86.3, 69.6, 44.8, 24.8, 6.9, 13.1, 5.6, 3.9, 10.5, 6.5
Timepoint
Hydroxychloroquine
Glucocorticoids
MPAA
Azathioprine
Cyclophosphamide
Methotrexate
Chloroquine
Tacrolimus
Rituximab
Belimumab

DM=disease modification, MPAA=mycophenolic acid.

ACKNOWLEDGMENTS

This study was sponsored by Johnson & Johnson. Medical writing support was provided by Priyanka Bhosale, PhD; poster layout and graphic design support was provided by Samita Warang, both of SIRO Medical Writing Pvt. Ltd., India (funded by Johnson & Johnson).

DISCLOSURES

ACOSM: Served as a speaker for AstraZeneca and GSK. **MFU-G:** Received grant support from Janssen; received consulting and/or speaking fees from AstraZeneca, Ferrer, GSK, Novartis and Tecnofarma. **FZ, AO:** Employment—Johnson & Johnson, stock ownership—Johnson & Johnson. **SB-M, DF-A, OM, LH, ESPS, LM, PA, VS, LG, MES, MCB, MLM, GNG, MAC, ACR, JMS, OAM, ALBPD, LCAA, EFB, EB, ETdR-N, IGH, MM, GAM, LGE, CAC, GQ-L, CETG, JMMP, RES-B, MPC, EM-N, YJ-V, IG-V, ROH, JAEV, IAC, AP, JCC, ACQ, RA, AC, CP,** and **CD:** Declared no conflict of interest. **JEBC:** Supported by the Colombian Association of Rheumatology (ASOREUMA).

PRESENTED AT: AMERICAN COLLEGE OF RHEUMATOLOGY (ACR) CONVERGENCE; OCTOBER 24–29, 2025; CHICAGO, IL, USA.