0623

CLUSTER ANALYSIS OF SOCIOECONOMIC AND ENVIRONMENTAL DETERMINANTS MODIFYING ACTIVITY, CHRONICITY AND CLINICAL MANIFESTATIONS OF SYSTEMIC LUPUS ERYTHEMATOSUS IN THE GLADEL 2.0 COHORT

Marco Ulises Martínez Martínez*, Karen Roberts, Rosana Quintana, Marina Scolnik, Carmen Funes Soaje, Paula Alba, Verónica Saurit, Mercedes Argentina García, Guillermo A. Berbotto, Verónica Bellomio, Eduardo Mario Kerzberg, Graciela Noemí Gómez, Cecilia Nora Pisoni, Vicente Juárez, Ana Malvar, Nilzio Antonio da Silva, Odirlei Andre Monticielo, Henrique Ataide Mariz, Francinne Machado Ribeiro, Eduardo F. Borba, Eloisa Bonfa, Edgard Torres dos Reis-Neto, Iris Guerra Herrera, Loreto Massardo, Gustavo Aroca Martínez, Lorena Gómez Escorcia, Carlos A. Cañas, Gerardo Quintana-Lopez, Carlos Enrique Toro Gutiérrez, Mario Javier Moreno Alvarez, Miguel Ángel Saavedra, Margarita Portela Hernández, Hilda Fragoso-Loyo, Luis H. Silveira, Ignacio García de la Torre, Carlos Abud-Mendoza, Jorge A. Esquivel Valerio, Isabel Acosta Colman, Astrid Paats, Claudia Mora Trujillo, Manuel F. Ugarte-Gil, Armando Calvo Quiroz, Roberto Muñoz Louis, Martín Rebella, Álvaro Danza, José A. Gómez-Puerta, Federico Zazzetti, Ashley Orillion, Guillermo Pons-Estel, Ingris Peláez-Ballestas, on behalf of Grupo Latino Americano De Estudio de Lupus (GLADEL), Study Group of PANLAR

*Presenting author

BACKGROUND

- Systemic lupus erythematosus (SLE) is a complex autoimmune disease with heterogeneous clinical manifestations
- Evidence from high-income countries suggests that socioeconomic status (SES) and environmental factors influence SLE outcomes, yet data from Latin America remain limited

OBJECTIVE

- The objective of this research was to evaluate, across three levels (individual, regional, and national), whether socioeconomic and environmental factors impact the clinical presentation of SLE in the GLADEL 2.0 cohort

METHODS

- We analyzed data from 1,083 patients with SLE enrolled in the GLADEL 2.0 cohort across 10 Latin American countries
 - All patients fulfilled the 1982/1997 American College of Rheumatology (ACR) or 2012 Systemic Lupus International Cooperating Clinics (SLICC) classification criteria
- Clinical outcomes were disease activity (Systemic Lupus Erythematosus Disease Activity Index 2000 [SLEDAI-2K]), organ involvement (renal, hematologic, cutaneous, musculoskeletal), and organ damage (SLICC/ACR damage index; SDI)
- Variables were structured into three levels: Level 1 (individual), Level 2 (state/province), and Level 3 (national). Multilevel analyses included sociodemographic variables, environmental exposure to air pollutants ($PM_{2.5}$, PM_{10}, NO_2), and contextual indicators (income, unemployment, distribution of income or wealth through the Gini index, CO_2 emissions)
- Statistical analysis included cluster analysis (Ward's method) and principal component analysis (PCA)

RESULTS

- We identified five distinct clusters of patients with SLE, characterized by unique combinations of clinical severity, patient-reported outcomes (PROs), SES, and environmental exposures
 - Number of patients per cluster were: cluster 1 (n=285); cluster 2 (n=29); cluster 3 (n=74); cluster 4 (n=433); cluster 5 (n=262)
- Cluster 1 comprised mainly Argentinian (n=223, 78.2%) and Uruguayan (n=49, 17.2%) patients
- All patients in cluster 2 were from the Dominican Republic (n=29; 100%)
- All patients in cluster 3 were from Peru (n=74, 100%)
- Cluster 4 comprised mainly Brazilian (n=182, 42%), Colombian (n=137, 31.6%), and Paraguayan (n=57, 13.2%) patients
- Cluster 5 comprised mainly Mexican (n=176, 67.2%), Argentinian (n=55, 21%), and Chilean (n=31, 11.8%) patients
- Heatmaps illustrate select sociodemographic and clinical features (**Figures 1, 2**), environmental factors (**Figure 2**), and patient reported outcomes (**Figure 3**) across the five clusters
- PCA showed that clinical manifestations, disease activity, and chronicity were different across the five clusters (**Figure 4**)

CONCLUSIONS

- Socioeconomic and environmental factors at individual, regional, and national levels influence the cumulative clinical manifestations and patient-reported outcomes in SLE
- Our findings underscore the role of social vulnerability and environmental stressors as potential modifiers of SLE phenotype in Latin America, highlighting the need for context-sensitive interventions and policies to mitigate health inequities

ACKNOWLEDGMENTS

This study was sponsored by Johnson & Johnson. Medical writing support was provided by Jyotsana Dixit; poster layout and graphic design support was provided by Sandeep Chavan, both from SIRO Medical Writing Pvt Ltd, India (funded by Johnson & Johnson).

DISCLOSURES

MS: Received speaker fees, advisory board fees, and/or grants from GSK, AstraZeneca, Janssen, Roche, and Pfizer. **GP-E:** Received grants and consulting fees from and participated as a speaker, as an advisor, and/or on a steering committee for AstraZeneca, Boehringer Ingelheim, GSK, Janssen, Novartis, Pfizer, RemeGen, Sanofi, and Werfen Diagnostics. **JAG-P:** Speaker for AstraZeneca, GSK, Lilly, Otsuka, Pfizer. **MFU-G:** Received grant support from Janssen; received consulting and/or speaking fees from AstraZeneca, Ferrer, GSK, Novartis and Tecnofarma. **MUMM, KR, RQ, CFS, PA, VS, MAG, GAB, VB, EMK, GNG, CNP, VJ, AM, NAdS, OAM, HAM, FMR, EFB, EB, ETdR-N, LM, GAM, CAC, GQ-L, CETG, MJMA, MAS, MPH, HF-L, LHS, IGdIT, CA-M, JAEV, IAC, AP, CMT, ACQ, RML, MR, AD, IPB:** Declared no conflicts of interest. **FZ, AO:** Employment—Johnson & Johnson, stock ownership—Johnson & Johnson.

FIGURE 1. Sociodemographic and clinical characteristics across clusters

Sociodemographic, clinical, and socioeconomic variables

Group	Variable	Cluster 1	Cluster 2	Cluster 3	Cluster 4	Cluster 5
A&E	Age at diagnosis (years)	29.5 (11.5)	29.7 (8.5)	33.1 (13.2)	27.9 (10.5)	29.4 (12.7)
A&E	Education (years)	14.5 (4.15)	13.7 (3.43)	13.8 (3.32)	12.6 (3.62)	13 (3.75)
Sx	Female	88.4%	96.6%	87.8%	91.2%	87.8%
Ethnic	Afro-Latin American	1.1%	100%	0%	13.4%	0%
Ethnic	Indigenous	1.8%	0%	0%	0.9%	0%
Ethnic	Mixed	31.9%	0%	95.9%	65.6%	97.3%
Ethnic	Others	0.4%	0%	0%	0.2%	0%
Ethnic	White	64.2%	0%	4.1%	19.4%	2.7%
Marital	Married/Concubine	33.3%	37.9%	54.1%	44.3%	44.7%
Marital	Separated/Divorced	8.1%	6.9%	2.7%	4.4%	6.1%
Marital	Single	47%	55.2%	40.5%	47.6%	47.7%
Marital	Widowed	1.4%	0%	2.7%	1.2%	0.8%
SES	High	1.4%	0%	1.4%	2.3%	4.2%
SES	Low	4.6%	27.6%	14.9%	14.3%	10.3%
SES	Middle	48.4%	20.7%	35.1%	33.3%	24.8%
SES	Middle high	24.6%	0%	36.5%	15.5%	22.9%
SES	Middle low	20.4%	51.7%	12.2%	32.6%	35.9%
Medical	Full coverage	81.4%	6.9%	86.5%	50.1%	32.4%
Medical	Partial coverage	2.8%	89.7%	8.1%	6.7%	32.1%
Medical	Without coverage	14%	3.4%	5.4%	41.6%	34.7%
Occupation	Full-time work	54.7%	3.4%	47.3%	33.3%	29.8%
Occupation	Partial-time work	15.4%	27.6%	16.2%	9%	21%
Occupation	Retired	6.3%	0%	2.7%	4.2%	2.7%
Occupation	Student	7%	27.6%	4.1%	9%	13.4%
Occupation	Unemployed (looking for job)	5.6%	10.3%	18.9%	16.9%	17.2%
Occupation	Unemployed due to illness	5.6%	27.6%	9.5%	22.2%	10.7%
Occupation	Unknown	5.3%	3.4%	1.4%	5.5%	5.3%

RSV: 1, 0, −1

A&E=age and education, RSV=relative standardized values, SES=socioeconomic status, Sx=sex.

FIGURE 2. Socioeconomic and environmental characteristics across clusters
Mean ± standard deviation for selected sociodemographic, environmental, and clinical indicators across the five clusters identified in the study cohort

Socioeconomic and environmental GLV

Group	Variable	Cluster 1	Cluster 2	Cluster 3	Cluster 4	Cluster 5
DU	Population density	1750 (874)	9850 (0)	3880 (0)	4320 (2860)	2990 (2550)
Environmental/Climate	CO2 emission	3.72 (0.64)	2.07 (0)	1.81 (0)	2.09 (0.64)	3.81 (0.45)
Environmental/Climate	DNI	1970 (123)	1860 (0)	1090 (0)	1520 (287)	2110 (184)
Environmental/Climate	GHI	1780 (69.9)	2020 (0)	1700 (0)	1810 (167)	2060 (132)
Environmental/Climate	NO_2_concentration	16.9 (4.39)	1.66 (0)	48.7 (0)	20.1 (7.9)	30.9 (14.4)
Environmental/Climate	PM_{10} concentration	29.8 (12.5)	79.1 (0)	71.3 (0)	32.4 (10.8)	47.5 (10.5)
Environmental/Climate	$PM_{2.5}$_concentration	13.6 (6.1)	56 (0)	28.6 (0)	15 (2.81)	22.4 (6.93)
HS	Physicians/1000 inhabitants	9.96 (5.83)	6.18 (0)	1.65 (0)	3.91 (1.58)	3.17 (1.13)
Socioeconomic	AMI USD/month	644 (317)	940 (0)	310 (0)	498 (237)	470 (252)
Socioeconomic	GDI	0.999 (0.01)	1.03 (0)	0.952 (0)	0.998 (0)	0.982 (0.01)
Socioeconomic	Gini index	0.400 (0.03)	0.375 (0)	0.400 (0)	0.473 (0.06)	0.392 (0.04)
Socioeconomic	HDI	0.830 (0.08)	0.781 (0)	0.765 (0)	0.752 (0.05)	0.818 (0.04)
Socioeconomic	Unemployment rate	8.07 (0.95)	5.7 (0)	6.6 (0)	8.18 (2.69)	5.29 (2.88)

RSV: 1, 0, −1

AMI USD=average monthly income in US dollars, DNI=direct normal irradiance (kWh/m²/year), DU=demographic and urban, GDI=Gender Development Index, GHI=global horizontal irradiance (kWh/m²/year), GLV=global level variables, HDI=Human Development Index, HS=health system, RSV=relative standardized values.

FIGURE 3. Patient reported outcomes across clusters
Clusters displayed distinct profiles across organ damage, disease activity, and patient-reported outcomes

Outcome	Cluster 1	Cluster 2	Cluster 3	Cluster 4	Cluster 5
SLICC*	1.0 (1.4)	0.7 (0.9)	1.7 (1.9)	1.1 (1.4)	1.1 (1.7)
SLEDAI**	7.7 (6.2)	13.0 (9.2)	9.3 (8.3)	9.8 (7.8)	10.3 (8.2)
Planning**	80.9 (22.1)	88.6 (7.9)	79.8 (17.4)	73.6 (25.8)	76.0 (23.1)
Physical Function**	79.3 (19.6)	89.7 (10.3)	75.8 (20.3)	74.4 (21.8)	77.3 (19.0)
Physical Burden**	74.9 (19.2)	85.6 (18.1)	76.8 (18.1)	68.0 (22.7)	73.1 (21.2)
Pain**	76.9 (21.7)	89.2 (10.6)	74.5 (17.4)	70.5 (23.3)	75.0 (20.6)
Intimacy*	78.1 (26.1)	88.1 (17.7)	75.6 (28.4)	73.1 (27.7)	76.6 (27.1)
Fatigue*	67.2 (25.4)	79.7 (15.5)	71.0 (20.7)	65.1 (24.3)	65.0 (24.4)
Emotional Burden**	71.7 (22.1)	79.7 (15.0)	68.8 (20.3)	64.9 (22.7)	66.3 (22.3)
Deterioration of Activities**	30.9 (25.6)	7.9 (10.3)	32.1 (22.9)	38.6 (27.0)	36.0 (25.1)
Cognition**	73.0 (25.5)	87.3 (20.9)	75.0 (19.9)	69.5 (25.5)	70.7 (25.5)

RSV: 1.00, 0.75, 0.50, 0.25, 0.00

RSV=relative standardized values, SLEDAI=Systemic Lupus Erythematosus Disease Activity Index, SLICC=Systemic Lupus International Collaborating Clinics.

FIGURE 4. Three-dimensional representation of the first three principal components
Five clusters of SLE patients derived from socioeconomic and environmental variables

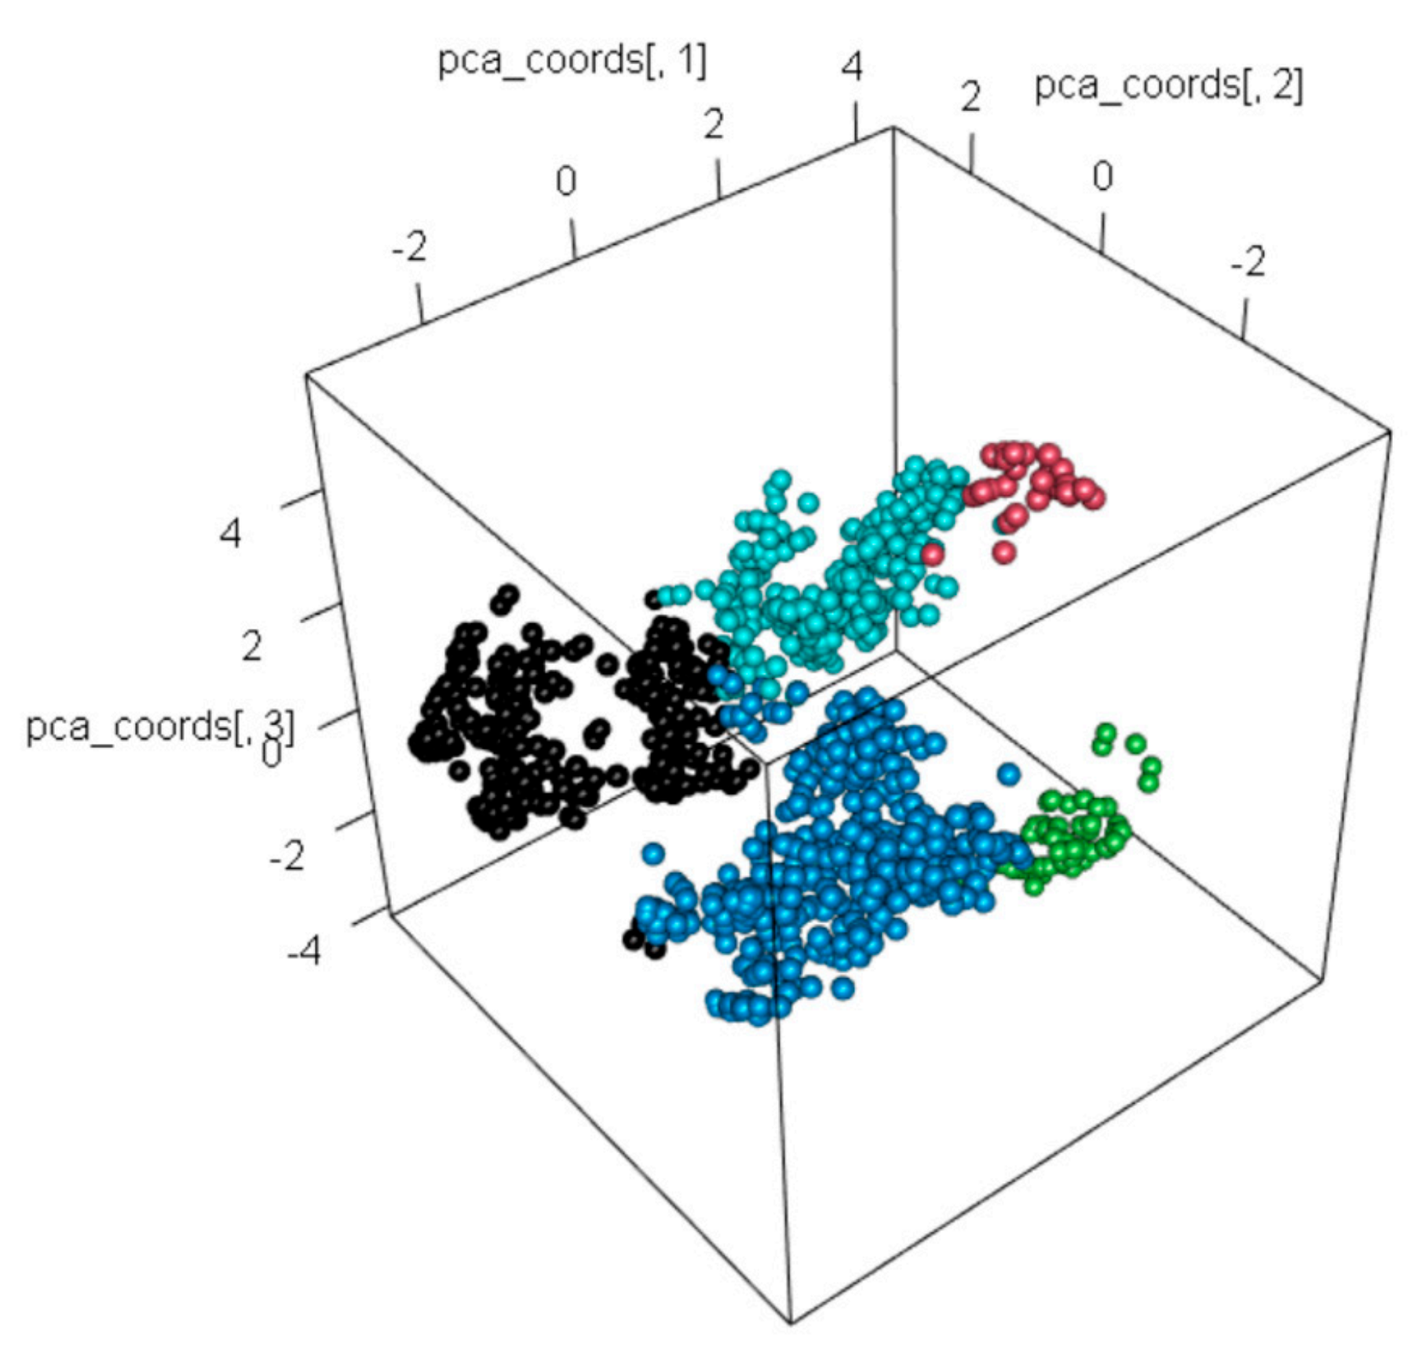
pca_coords[, 1]
pca_coords[, 2]
pca_coords[, 3]
-2
0
2
4
2
0
-2
4
2
0
-2
-4

PCA=principal components analysis, SLE=systemic lupus erythematosus.

PRESENTED AT: AMERICAN COLLEGE OF RHEUMATOLOGY (ACR) CONVERGENCE 2025; CHICAGO, IL, USA; OCTOBER 24–29, 2025.