0596

EFFECT OF GENDER AND FOLLOW-UP TIME IN DAMAGE ACCRUAL: DATA FROM A LATIN AMERICA LUPUS COHORT

Diana Fernández-Ávila, Rosana Quintana, Karen Roberts, Romina Nieto, Marina Scolnik, Carmen Funes Soaje, Cintia Otaduy, Verónica Saurit, Valeria Arturi, Guillermo A. Berbotto, Maria Constanza Bertolaccini, Eduardo Mario Kerzberg, María de los Angéles Gargiulo, Cecilia Nora Pisoni, Ana Carolina Ralle, Joaquín Martinez Serventi, Ana Carolina O. S. Montandon, Odirlei Andre Monticielo, Henrique Ataide Mariz, Laíssa Cristina Alves Alvino, Eduardo F. Borba, Emily Figueiredo Neves Yuki, Edgard Torres dos Reis-Neto, Iris Guerra Herrera, Milena Mimica Davet, Gustavo Aroca Martínez, Antonio Iglesias Gamarra, Carlos A. Cañas Davila, Gerardo Quintana-Lopez, Carlos Enrique Toro Gutiérrez, Mario Javier Moreno Alvarez, Olga Lidia Vera Lastra, Margarita Portela Hernández, Hilda Fragoso Loyo, Luis H. Silveira, Yelitza González Bello, Carlos Abud Mendoza, Jorge Antonio Esquivel-Valerio, Marcelo Barrios, Carolina Vázquez, Magaly Alva Linares, Manuel F. Ugarte-Gil, Armando Calvo Quiróz, Roberto Muñoz Louis, Carina Pizzarossa, Gonzalo Silveira, Federico Zazzetti, Ashley Orillion, Graciela S. Alarcon, Bernardo Pons-Estel, Guillermo Pons-Estel*, on behalf of Grupo Latino Americano De Estudio de Lupus (GLADEL)

*Presenting author

BACKGROUND

- Previous studies have shown that male gender is an independent predictor of organ damage in patients with systemic lupus erythematosus (SLE), particularly in the early stages of their disease[1,2]
- In the previous GLADEL cohort, male patients were found to have a higher SDI (SLICC/ACR Index) than their female counterparts, although this difference was not statistically significant[3]

OBJECTIVE

This study aimed to evaluate the impact of gender on organ damage (as assessed by SDI) in patients with SLE in the GLADEL 2.0 cohort

METHODS

STUDY POPULATION

- GLADEL 2.0 is an observational, multiethnic, multinational Latin American SLE cohort
- A total of 44 centers from 10 Latin American countries enrolled patients ≥18 years of age who met the 1982/1997 ACR (American College of Rheumatology) and/or the 2012 SLICC (Systemic Lupus International Collaborating Clinics) classification criteria

STATISTICAL ANALYSIS

- Continuous variables were summarized as medians (interquartile ranges) and categorical variables as frequencies (percentages)
- Logistic regression modeling was used to identify factors associated with the increase in the SDI between the baseline visit and the end of follow-up
 - *P* values <0.05 were considered statistically significant
- An interaction analysis between gender and time was performed using a mixed R model, with SDI as a dependent variable and gender (between-subjects) and time (within-subjects) as independent variables
 - A random intercept per participant was included to control for within-subject variability
- All analyses were descriptive and were done using R v4.40

RESULTS

- Of the 1081 patients enrolled in the GLADEL 2.0 cohort, 385 patients matched by gender were considered (**Table 1**)
 - 190 patients completed at least four annual visits and were included in these analyses
 - 70% of the patients were women and 30% were men
- No statistically significant differences in organ damage as measured by the SDI were found between male and female patients (*P*=0.563)
 - This trend remained constant throughout the follow-up period, with no significant interactions observed (*P*=0.904)
- However, the percentage of patients who had an increase in the SDI at the end of the follow-up period was higher among male versus female patients (88.6% vs 54.6%; **Figure 1A**)
 - The accrual of damage over the follow-up time increased significantly for both male and female patients
- The frequency of affected SDI domain was examined both at baseline and during follow-up (**Figure 1B**)
 - In general, males had a higher occurrence of ocular, cutaneous, and malignant involvement compared with females
 - Females had a higher frequency of gastrointestinal, pulmonary, peripheral vascular, and musculoskeletal involvement than males
- After adjusting for sociodemographic and clinical/immunologic characteristics, multivariate analysis found that disease duration (OR: 1.05; 95%CI: 1.0–1.1) and disease activity, as measured by the SLEDAI−2K (OR: 1.05; 95%CI: 1.0–1.1), were associated with a higher likelihood of an increase in SDI

TABLE 1:
Sociodemographic and clinical characteristics of patients with SLE by gender

	Male (n=113)	Female (n=272)	*P* value	Total (N=385)
Age at diagnosis, years, median [IQR]	27.6 [22.6–37.6]	26.2 [20.0–35.1]	0.129	26.5 [20.9–35.5]
Ethnic group, n (%)			0.204	
Afro-Latin American	6 (5.4)	26 (9.6)		32 (8.4)
Indigenous	2 (1.8)	1 (0.4)		3 (0.8)
Mestizo	82 (73.2)	178 (65.7)		260 (67.9)
White	22 (19.6)	65 (24.0)		87 (22.7)
Other	0 (0)	1 (0.4)		1 (0.3)
Socioeconomic status, n (%)			0.287	
High	3 (2.7)	6 (2.3)		9 (2.4)
Upper middle	21 (18.9)	40 (15.3)		61 (16.4)
Middle	49 (44.1)	94 (35.9)		143 (38.3)
Lower middle	28 (25.2)	94 (35.9)		122 (32.7)
Low	10 (9.0)	28 (10.7)		38 (10.2)
Missing	2 (1.8)	10 (3.7)		12 (3.1)
Medical coverage, n (%)			0.154	
Total coverage	62 (55.9)	142 (53.0)		204 (53.8)
Partial coverage	20 (18.0)	33 (12.3)		53 (14.0)
Without coverage	29 (26.1)	93 (34.7)		122 (32.2)
Missing	2 (1.8)	4 (1.5)		6 (1.6)
SLICC at baseline, median [IQR]	0 [0–1.00]	0 [0–1.00]	0.56	0 [0–1.00]
Disease duration, years, median [IQR]	2.35 [0.192–8.92]	2.91 [0.562–9.82]	0.286	2.70 [0.452–9.50]
Use of glucocorticoids at baseline, n (%)	64 (97.0)	158 (95.8)	1.000	222 (96.1)
Use of antimalarial at baseline, n (%)	64 (97.0)	162 (97.0)	1.000	226 (97.0)
Type of lupus, n (%)			0.838	
1=Patient with SLE, no history of lupus nephritis	31 (27.4)	82 (30.1)		113 (29.4)
2=Patient with SLE, with prevalent inactive lupus nephritis	21 (18.6)	55 (20.2)		76 (19.7)
3=Patient with SLE, with active prevalent lupus nephritis	28 (24.8)	67 (24.6)		95 (24.7)
4=Patient with SLE, with incident (active) lupus nephritis	33 (29.2)	68 (25.0)		101 (26.2)

IQR=Interquartile range; SLE=Systemic lupus erythematosus; SLICC=Systemic Lupus International Collaborating Clinics.

FIGURE 1:
(A) Mean SLICC/ACR score and (B) frequency of manifestations by SDI area by gender and follow-up time

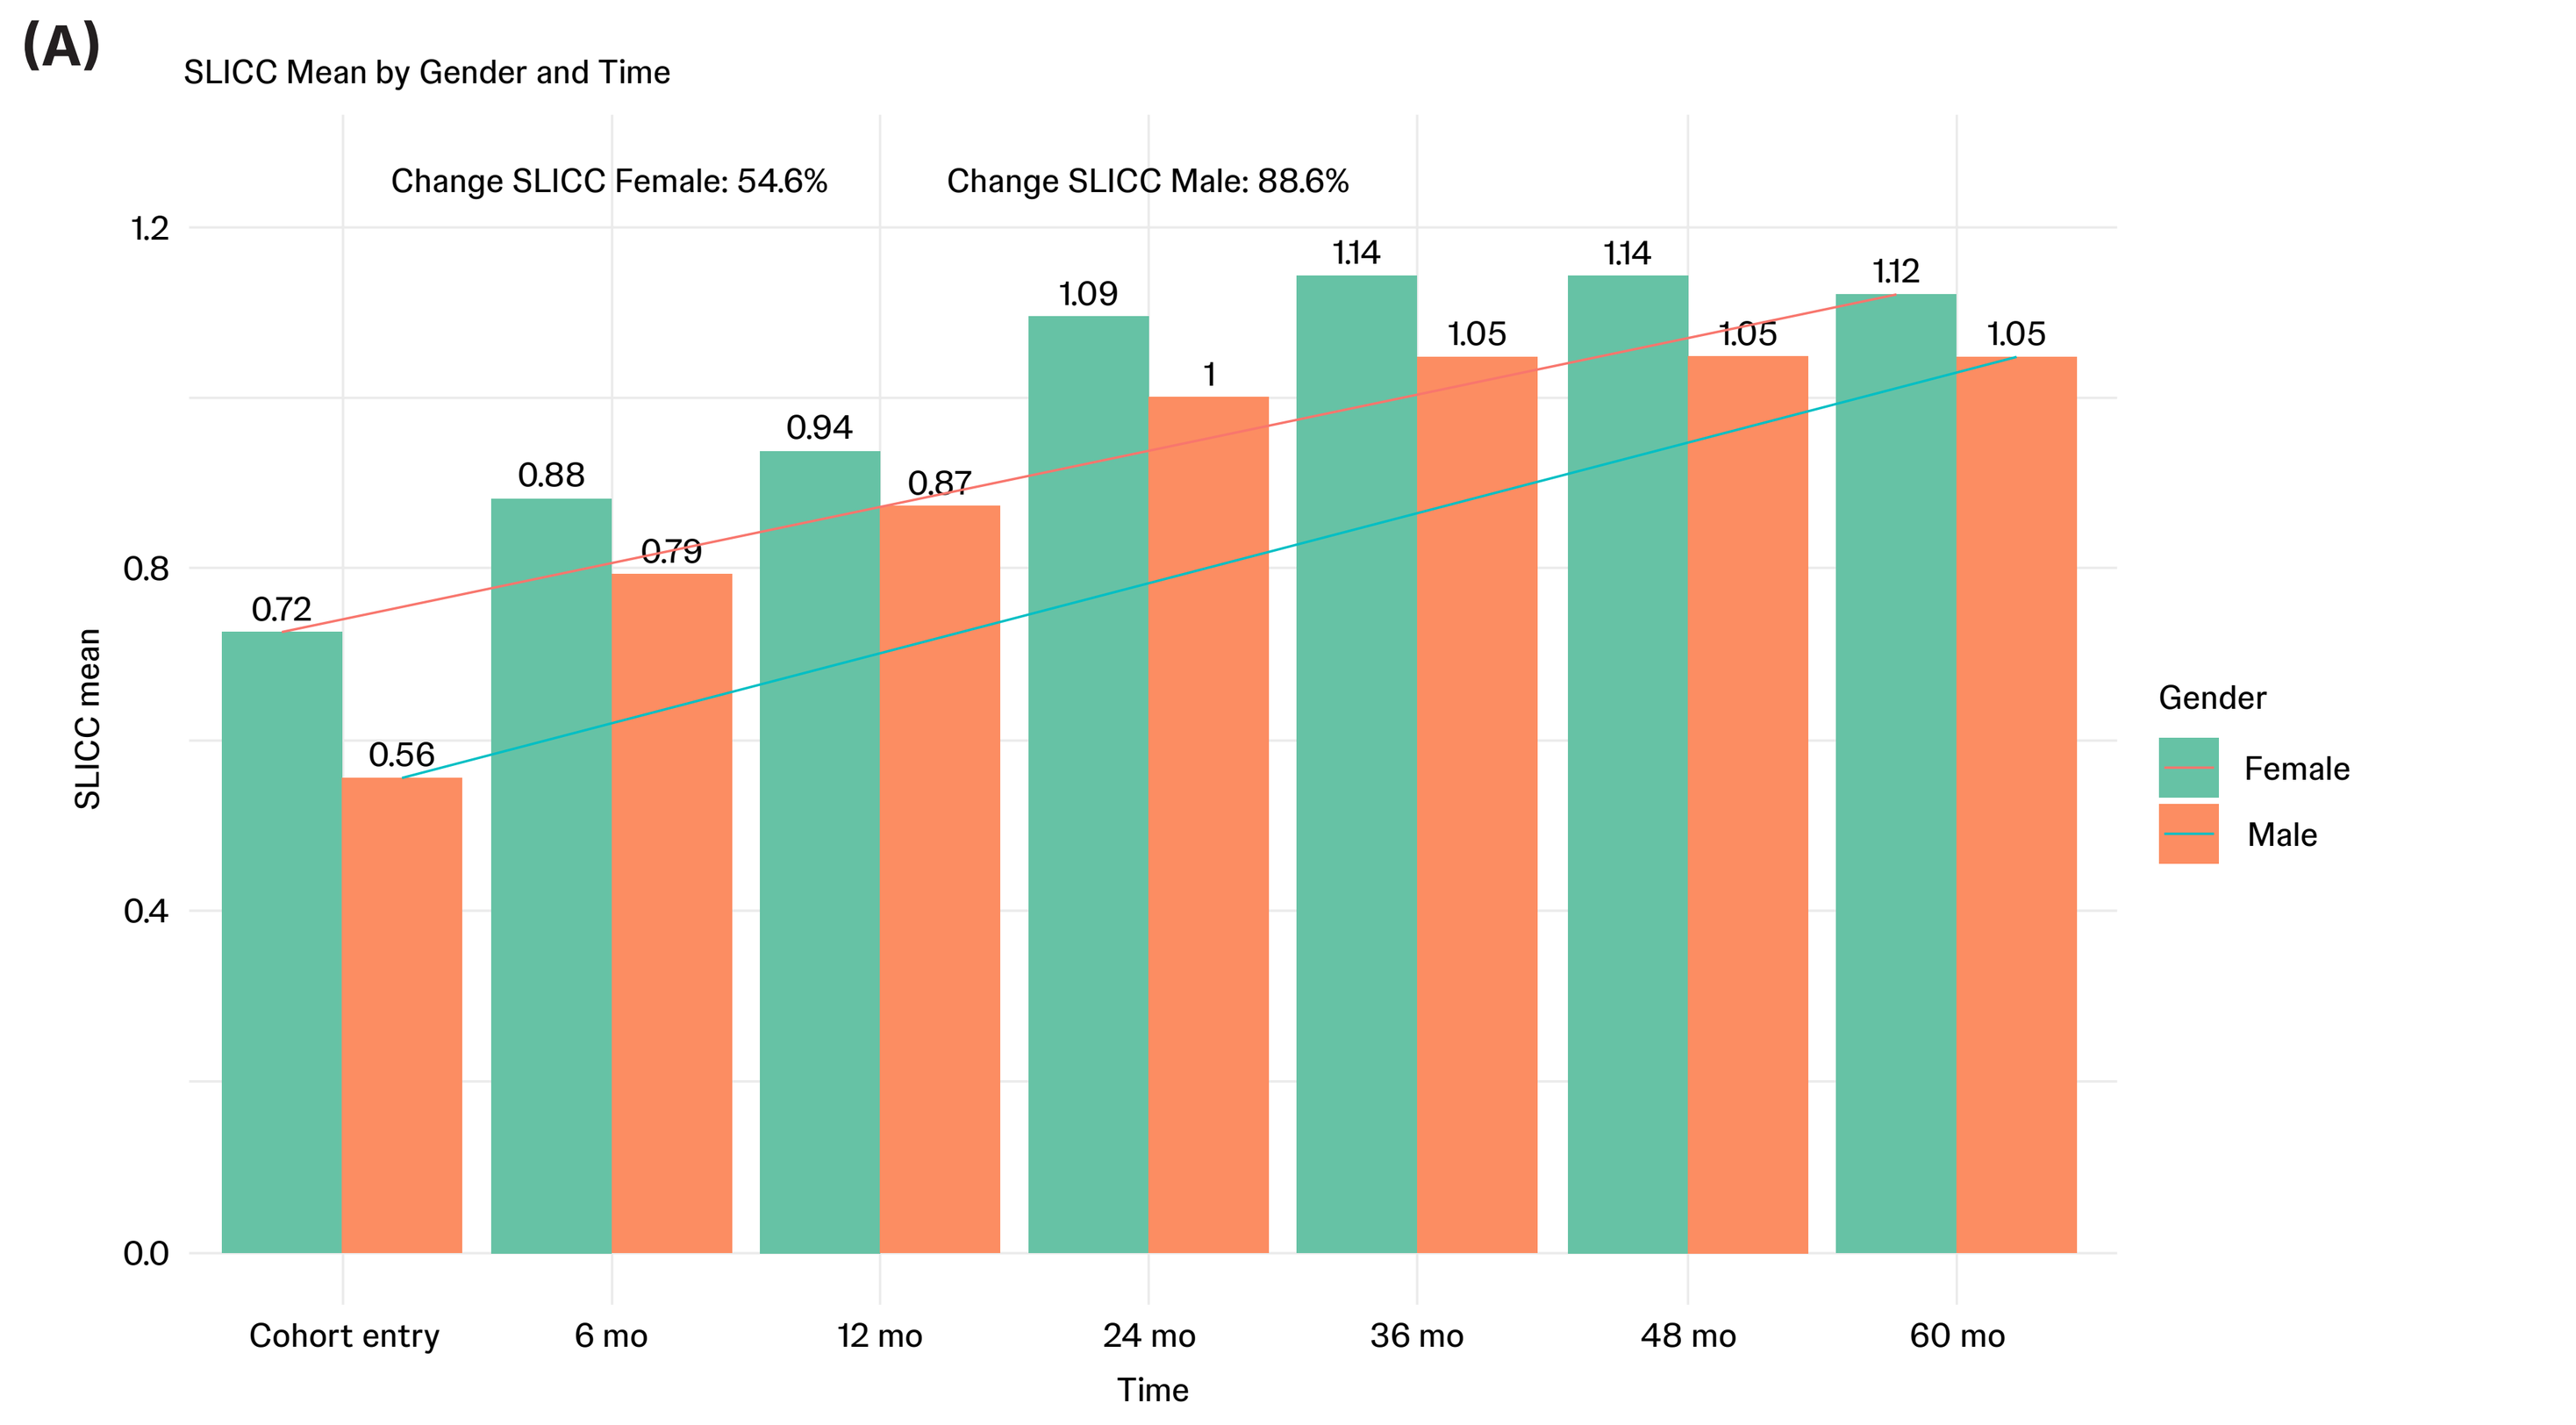
(A)
SLICC Mean by Gender and Time
Change SLICC Female: 54.6%
Change SLICC Male: 88.6%
SLICC mean
0.72
0.56
0.88
0.79
0.94
0.87
1.09
1
1.14
1.05
1.14
1.05
1.12
1.05
Cohort entry
6 mo
12 mo
24 mo
36 mo
48 mo
60 mo
Time
Gender
Female
Male

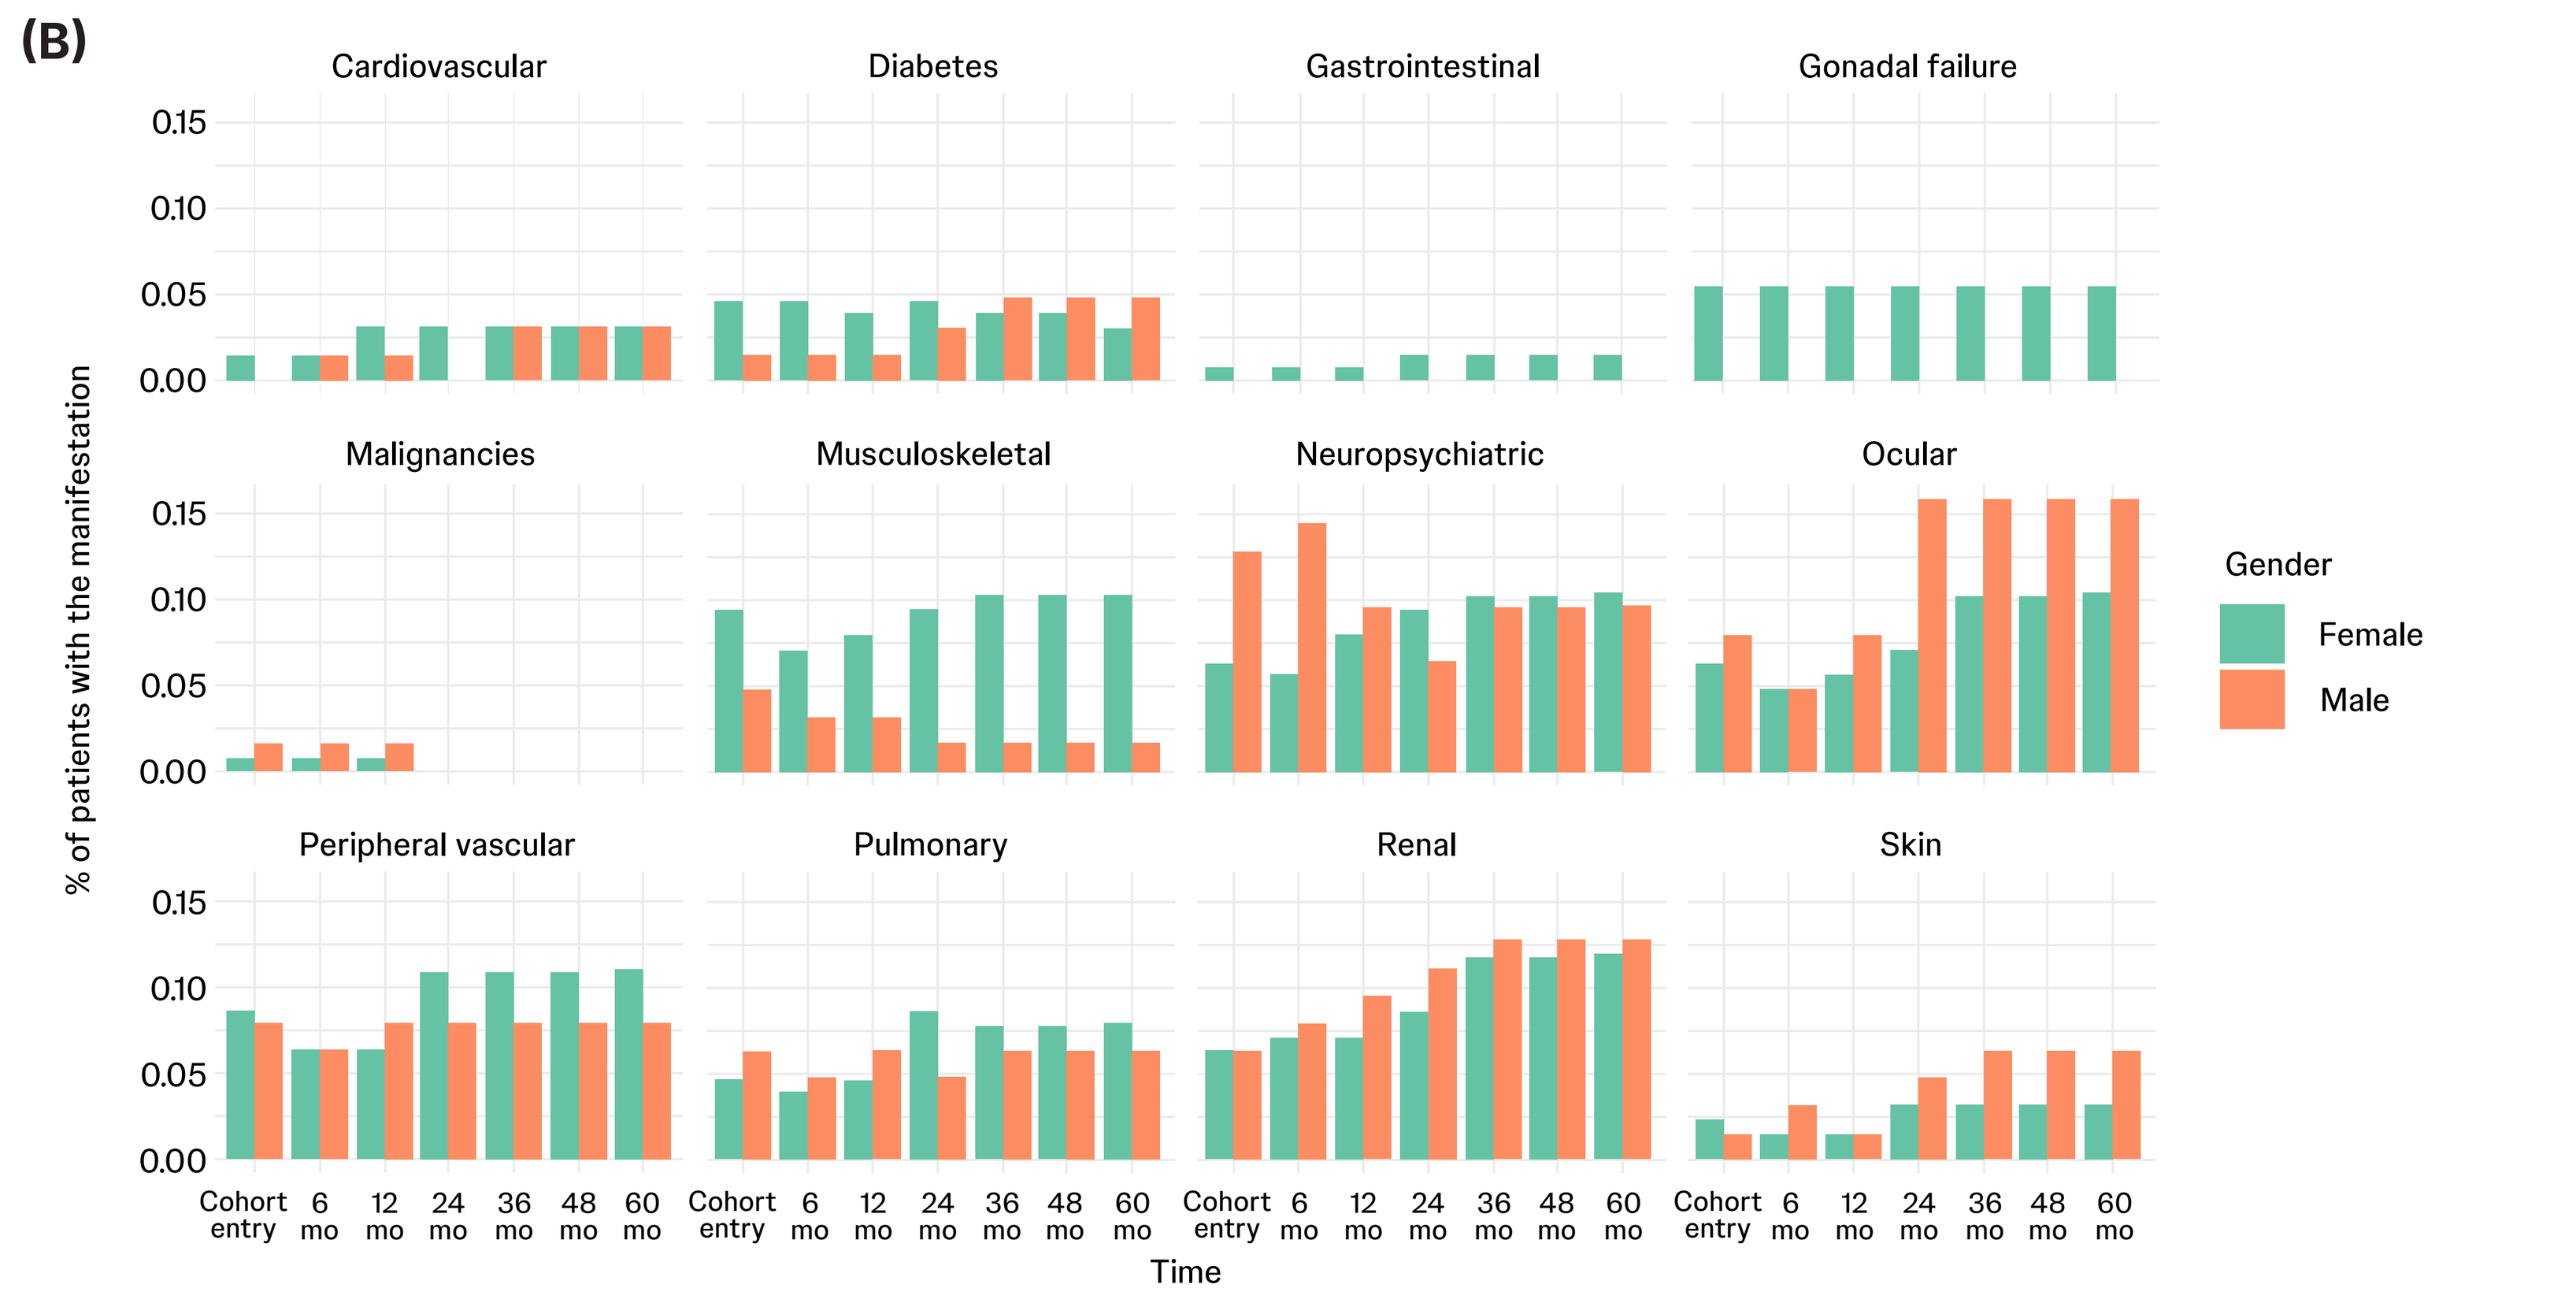
(B)
Cardiovascular
Diabetes
Gastrointestinal
Gonadal failure
Malignancies
Musculoskeletal
Neuropsychiatric
Ocular
Peripheral vascular
Pulmonary
Renal
Skin
% of patients with the manifestation
Time
Gender
Female
Male

ACR=American College of Rheumatology; mo=Month; SDI=Systemic Lupus International Collaborating Clinics/American College of Rheumatology Damage Index; SLICC=Systemic Lupus International Collaborating Clinics.

CONCLUSIONS

- Although male gender was not associated with an increased risk of damage accrual, the proportion of patients whose SDI increased was higher in males than females
- Organ damage during follow-up was significantly increased in both genders
- As patients are followed closely for a longer duration, the impact of gender on damage accrual may become more apparent

REFERENCES

1. Bruce IN, et al. *Ann Rheum Dis.* 2025:74:1706–13.
2. Andrade RM, et al. *Arthritis Rheum*. 2007;56:622–30.
3. MA García, et al. *Lupus*. 2005;14;938–046.

ACKNOWLEDGMENTS

This study was sponsored by Johnson & Johnson. Medical writing support was provided by Namiko Abe, PhD, of Joulé Clinical, and was funded by Johnson & Johnson. Layout design and reformatting for this encore presentation was provided by Sandeep Chavan of SIRO Medical Writing Pvt. Ltd., Thane, Maharashtra, India.

DISCLOSURES

D. Fernández-Ávila, R. Quintana, K. Roberts, R. Nieto, C. Funes Soaje, C. Otaduy, V. Saurit, V. Arturi, G. A. Berbotto, M. C. Bertolaccini, E. M. Kerzberg, M. de los Á. Gargiulo, C. Pisoni, A. C. Ralle, J. Martinez Serventi, O. A. Monticielo, H. A. Mariz, L. C. A. Alvino, E. F. Borba, E. F. N. Yuki, E. T. dos Reis-Neto, I. G. Herrera, M. Mimica Davet, G. Aroca Martínez, A. I. Gamarra, C. A. Cañas, G. Quintana-Lopez, C. E. T. Gutiérrez, M. J. Moreno Alvarez, O. L. Vera-Lastra, M. Portela Hernández, H. Fragoso Loyo, L. H. Silveira, Y. González Bello, C. Abud Mendoza, J. A. Esquivel-Valerio, M. Barrios, C. Vázquez, M. A. Linares, A. C. Quiróz, R. M. Louis, C. Pizzarossa, G. Silveira, and **G. S. Alarcón:** Declares no conflict of interest. **M. Scolnik:** Received speaker fees and/or served in advisory roles for GSK, AstraZeneca, Janssen, Roche, and Pfizer; and has received grants from GSK, AstraZeneca, Janssen, Roche, and Pfizer. **A. C. O. S. Montandon:** Received support from GSK and AstraZeneca. **M. F. Ugarte-Gil:** Received grant support from Janssen; received consulting and/or speaking fees from AstraZeneca, Ferrer, GSK, Novartis and Tecnofarma. **F. Zazzetti, A. Orillion:** Employees of Johnson & Johnson and may hold stocks or stock options in the company. **B. Pons-Estel:** Served as a speaker and/or advisor for AstraZeneca, GSK, and Janssen. **G. Pons-Estel:** Served as a speaker, advisor, and/or on steering committees for AstraZeneca, Boehringer Ingelheim, GSK, Janssen, Novartis, Pfizer, RemeGen, Sanofi, and Werfen Diagnostics; and has received grants and consulting fees from these same companies. Previously presented at EULAR 2025; Barcelona, Spain; June 11–14, 2025.

PRESENTED AT: AMERICAN COLLEGE OF RHEUMATOLOGY (ACR) CONVERGENCE 2025; CHICAGO, IL, USA; OCTOBER 24–29, 2025.