0621

RISK FACTORS FOR PULMONARY MANIFESTATIONS IN GLADEL 2.0, A SYSTEMIC LUPUS ERYTHEMATOSUS LATIN AMERICAN COHORT

Luis Javier Cajas Santana*, Sebastián Molina Ríos, Lucia Hernández, Romina Nieto, Gricel Maita Romero, Nidia Noemí Merás, Veronica Gabriela Savio, Verónica Saurit, Valeria Arturi, Boris Kisluk, Verónica Bellomio, Eduardo Mario Kerzberg, Nicolás Perez, Cecilia Nora Pisoni, Vicente Juárez, Joaquín Martinez Serventi, Nilzio Antonio da Silva, Odirlei Andre Monticielo, Mariana Souza Pessoa de Luna, Laíssa Cristina Alves Alvino, Eduardo F. Borba, Luciana Parente Costa Seguro, Edgard Torres dos Reis-Neto, Iris Guerra Herrera, Milena Mimica, Gustavo Aroca Martínez, Valentina Pérez jiménez, Carlos A. Cañas, Gerardo Quintana-Lopez, Carlos Enrique Toro Gutiérrez, Rafael Ignacio López Martínez, Miguel Ángel Saavedra, Margarita Portela Hernández, Hilda Fragoso-Loyo, Luis H. Silveira, Yelitza C. González-Bello, Carlos Abud-Mendoza, Jorge A. Esquivel Valerio, Patricia Langjahr, Astrid Paats, Claudia Mora-Trujillo, Victor R. Pimentel-Quiroz, Carlos Bedia, Teresandris Polanco, Martín Rebella, Álvaro Danza, Federico Zazzetti, Ashley Orillion, Guillermo Pons-Estel, Graciela S. Alarcón, on behalf of Grupo Latino Americano De Estudio de Lupus (GLADEL), Study Group of PANLAR

*Presenting author

BACKGROUND

- Systemic Lupus Erythematosus (SLE) is a multisystemic autoimmune disease
- Pleuropulmonary manifestations, including pleural effusion, interstitial lung disease, pulmonary hypertension, shrinking lung syndrome, and alveolar hemorrhage have been described[1,2]; however, data on their frequency and the risk factors associated with their occurrence are scarce in the Latin American SLE population
- A better understanding of pleuropulmonary manifestations in this population is crucial due to the risk of severe and irreversible damage, and their direct impact on morbidity, mortality, and patient quality of life

OBJECTIVE

This study aimed at evaluating risk factors for pleuropulmonary manifestations among patients with SLE in the GLADEL 2.0 cohort

METHODS

- Patients from the Latin American GLADEL 2.0 cohort who met the American College of Rheumatology/European Alliance for Rheumatology (ACR/EULAR) 2019 and/or the Systemic Lupus International Cooperating Clinics (SLICC) SLE classification criteria were included
- Patients with pleuropulmonary manifestations (pleural effusion, interstitial lung disease, pulmonary hypertension, shrinking lung syndrome, alveolar hemorrhage) were identified and compared to those without them. Patients with polyautoimmunity, where the manifestation could have another explanation, were excluded
- Univariate analyses were conducted to determine the possible associations of each pleuropulmonary manifestation, using data recorded prior to the report of the pleuropulmonary manifestation
- Subsequently, multivariate logistic regression analyses were performed, adjusting for age, gender, and other variables found to be significant in the univariate analyses (p<0.05)

RESULTS

- From a total of 1,083 patients included in the GLADEL 2.0 cohort, 242 patients with pleural effusion, 38 with interstitial lung disease, 26 with pulmonary hypertension, 20 with shrinking lung syndrome, and 9 with alveolar hemorrhage were identified
- The factors associated with each manifestation, except for alveolar hemorrhage (due to small n), are summarized in **Table 1A & 1B** and **Table 2**
- Age at diagnosis, disease duration, history of renal involvement (including both active and inactive nephritis), and the degree of organ damage, assessed with the SLICC Damage Index (SDI), were frequently associated with pleuropulmonary manifestations **(Table 1A & 1B)**
- In the multivariate analysis, the association between the SLICC Damage Index and pleuropulmonary manifestations remained statistically significant for all complications, with odds ratios ranging from 1.35 to 1.56 **(Table 2)**
- Disease activity, measured with the SLE Disease Activity Index-2000 (SLEDAI-2K), was only found to be significant for pleural effusion (p=0.001)
- Significant associations between the presence of serositis and pulmonary hypertension (p=0.005) and SLS (p=0.001) were found

CONCLUSIONS

- Damage, as assessed by the SDI, was a consistent risk factor associated with pleuropulmonary manifestations among patients in the GLADEL 2.0 cohort
 - This risk factor has not been previously reported for the occurrence of pleuropulmonary manifestations in SLE
- These data highlight the importance of close monitoring for pleuropulmonary manifestations in patients with SLE who already have organ damage
- Additionally, serositis significantly increased the risk of developing pulmonary hypertension and shrinking lung syndrome in this cohort, consistent with data reported in previous studies
- Risk factors for alveolar hemorrhage could not be examined due to the small number of cases

TABLE 1A. Risk factors for pulmonary manifestations in GLADEL 2.0 cohort patients: univariate analysis (pleural disease, interstitial lung disease)

VARIABLE	TOTAL	NO COMPLICATION	COMPLICATION	*p* VALUE
Pleural disease (n)		791	242	
Age at diagnosis, years, median (IQR)	27.0 (20.6–35.4)	27.4 (20.8–36.1)	25.2 (19.8–34.0)	0.023
Lupus nephritis, n (%)				0.001
None, n (%)	430 (39.7)	338 (42.7)	92 (31.5)	
Lupus nephritis, active, n (%)	425 (39.2)	285 (36.0)	140 (47.9)	
Lupus nephritis, inactive, n (%)	228 (21.1)	168 (21.2)	20 (20.5)	
SLEDAI-2K, median (IQR)	3 (0–8)	2 (0–6)	6 (2–12)	0.001
24 hours proteinuria, median (IQR)	0.4 (0.1–1.7)	0.3 (0.1–1.1)	1.3 (0.3–3.3)	0.001
Anti dsDNA, positivity, n (%)	851 (78.6)	610 (77.1)	241 (82.5)	0.005
Interstitial lung disease (n)		1045	38	
Age at diagnosis, median (IQR)	27.0 (20.6–35.4)	26.9 (20.5–35.0)	30.7 (22.5–43.0)	0.050
Duration of disease, months, median (IQR)	68.0 (19.7–145.6)	67.8 (19.3–144.8)	101.7 (40.9–166.8)	0.041
Lupus nephritis, n (%)				0.015
None, n (%)	430 (39.7)	408 (39.0)	22 (57.9)	
Active, n (%)	425 (39.2)	418 (40.0)	7 (18.4)	
Inactive, n (%)	228 (21.1)	219 (21.0)	9 (23.7)	
SDI, median (IQR)	0 (0–1)	0 (0–1)	2 (1–3)	0.001
C3 complement decrease, n (%)	830 (76.6)	807 (77.2)	23 (60.5)	0.029
C4 complement decrease, n (%)	837 (77.3)	814 (77.9)	23 (60.5)	0.017

TABLE 1B. Risk factors for pulmonary manifestations in GLADEL 2.0 cohort patients: univariate analysis (pulmonary hyptertension, shrinking lung syndrome)

VARIABLE	TOTAL	NO COMPLICATION	COMPLICATION	*p* VALUE
Pulmonary hypertension		1057	26	
Duration of disease, months, median (IQR)	68 (19.7–145.6)	67.5 (19.3–141.5)	124 (64.5–242.1)	0.003
Lupus nephritis, n (%)				0.042
None, n (%)	430 (39.7)	422 (39.9)	8 (30.8)	
Active, n (%)	425 (39.2)	418 (39.5)	7 (26.9)	
Inactive, n (%)	228 (21.1)	217 (20.5)	11 (42.3)	
Serositis, n (%)	367 (33.9)	351 (33.2)	16 (61.5)	0.005
SDI, median (IQR)	0 (0–1)	0 (0–1)	2 (1–4)	0.001
Shrinking lung syndrome		1063	20	
Serositis, n (%)	367 (33.9)	355 (33.4)	12 (60)	0.017
SDI, median (IQR)	0 (0–1)	0 (0–1)	1 (1–3)	0.001

dsDNA=*double-stranded deoxyribonucleic acid,* ***IQR***=*interquartile range,* ***SLEDAI***=*SLE Disease Activity Index,* ***SDI***=*SLICC/ACR Damage Index.*

TABLE 2. Risk factors for pulmonary manifestations in GLADEL 2.0 cohort patients: Multivariate logistic regression

	OR (CI 95%)	*p* VALUE
Pleural disease		
Lupus nephritis, inactive	1.94 (1.03–3.64)	0.039
SLEDAI-2K	1.11 (1.07–1.14)	<0.001
Interstitial lung disease		
SDI	1.35 (1.03–1.77)	0.030
Pulmonary hypertension		
SDI	1.56 (1.30–1.93)	<0.001
Serositis	2.71 (1.12–6.55)	0.027
Shrinking lung syndrome		
Serositis	2.62 (1.01–6.80)	0.047
SDI	1.46 (1.17–1.82)	<0.001

CI=*confidence interval,* ***OR***=*odds ratio,* ***SLEDAI***=*SLE Disease Activity Index,* ***SDI***=*SLICC/ACR Damage Index.*

REFERENCES

1. Medlin JL, et al. *Semin Arthritis Rheum.* 2018;48(2):198–204.
2. Liu Y, et al. *Lupus.* 2023;32(11):1310–1319

ACKNOWLEDGMENTS

This study was sponsored by Johnson & Johnson. Medical writing support was provided by Kalpana Tilekar, PhD (SIRO Medical Writing Pvt. Ltd., India). Poster layout and graphic design support was provided by Ganesh Karpe (SIRO Medical Writing Pvt. Ltd., India) funded by Johnson & Johnson.

DISCLOSURES

LPCS: Received speaker fees from GSK and AstraZeneca. **GP-E:** Received grants and consulting fees from and participated as a speaker, as an advisor, and/or on a steering committee for AstraZeneca, Boehringer Ingelheim, GSK, Janssen, Novartis, Pfizer, RemeGen, Sanofi, and Werfen Diagnostics. **LJCS, SMR, LH, RN, GMR, NNM, VGS, VS, VA, BK, VB, EMK, NP, CNP, VJ, JMS, NAdS, OAM, MSPdL, LCAA, EFB, ETdR-N, IGH, MM, GAM, VPJ, CAC, GQ-L, CETG, RILM, MAS, MPH, HF-L, LHS, YCG-B, CA-M, JAEV, PL, AP, CM-T, VRP-Q, CB, TP, MR, AD**, and **GSA:** Declared no conflicts of interest. **FZ** and **AO:** Employment - Johnson & Johnson, stock ownership - Johnson & Johnson.

PRESENTED AT: AMERICAN COLLEGE OF RHEUMATOLOGY (ACR) CONVERGENCE OCTOBER 24–29, 2025; CHICAGO, IL, USA.